A Five-Year Study on Critical Results Compliance to The Joint Commission National Patient Safety Goal 02.03.01

(Report Critical Results of Test and Diagnostic Procedures on a Timely Basis)

Stacy R. Schultz, BA, Robert E. Watson, Jr., MD, PhD, Sherrie L. Prescott, RN, BSN, Gina K. Hesley, MD, Karl N. Krecke, MD
Department of Radiology
Mayo Clinic, Rochester, MN

Aim

To describe efforts over the past five years to improve Critical Results documentation and communication in a large radiology department and comply with The Joint Commission (TJC) National Patient Safety Goal (NPSG) 02.03.01

Methods

Each year of the 5-year study, one Critical Result was selected for measurement by the Radiology Safety and Accreditation Committee with approval by the Clinical Practice Committee.

Monthly, the quality improvement analyst queried the pertinent exam database tied to the selected Critical Result (e.g., Acute Pulmonary Embolism (PE) – CT Chest w/PE Protocol, Acute Deep Vein Thrombosis (DVT) – Extremity Veins Complete) along with specific report keywords (e.g., acute, positive). The analyst would review each of the cases looking for evidence of a Critical Result.

If a Critical Result was identified, the analyst would then verify that the radiologist:

1) Communicated the result to the ordering provider within 60 minutes from the time of the interpretation
2) Documented the name of the licensed care provider to whom the result was communicated
3) Documented the time of communication

The positive exam information (date, patient account number, time of interpretation, time of report finalization, time of communication, name of radiologist and resident) was transferred to an Excel spreadsheet.

Data was charted for:

1) Policy compliance to all three elements (Figure 2)
2) Communication vs documentation (Figure 3)

Critical Results Listing

Figure 1: Critical Results include but are not limited to:

- Leaking abdominal aortic aneurysm
- Acute aortic dissection (*measured*)
- Acute DVT (*measured*)
- Ectopic pregnancy
- Massive hemoperitoneum on CT or ultrasound
- Significant intracranial hemorrhage
- Pneumoperitoneum (no post-op)
- Pneumothorax, if unsuspected
- Tension pneumothorax
- Acute pulmonary embolism (*measured*)
- Acute spinal cord compression
- Unstable spine fracture
- Acute testicular/ovarian torsion
- Significant misplacement of tubes or catheters
- New perforated viscus

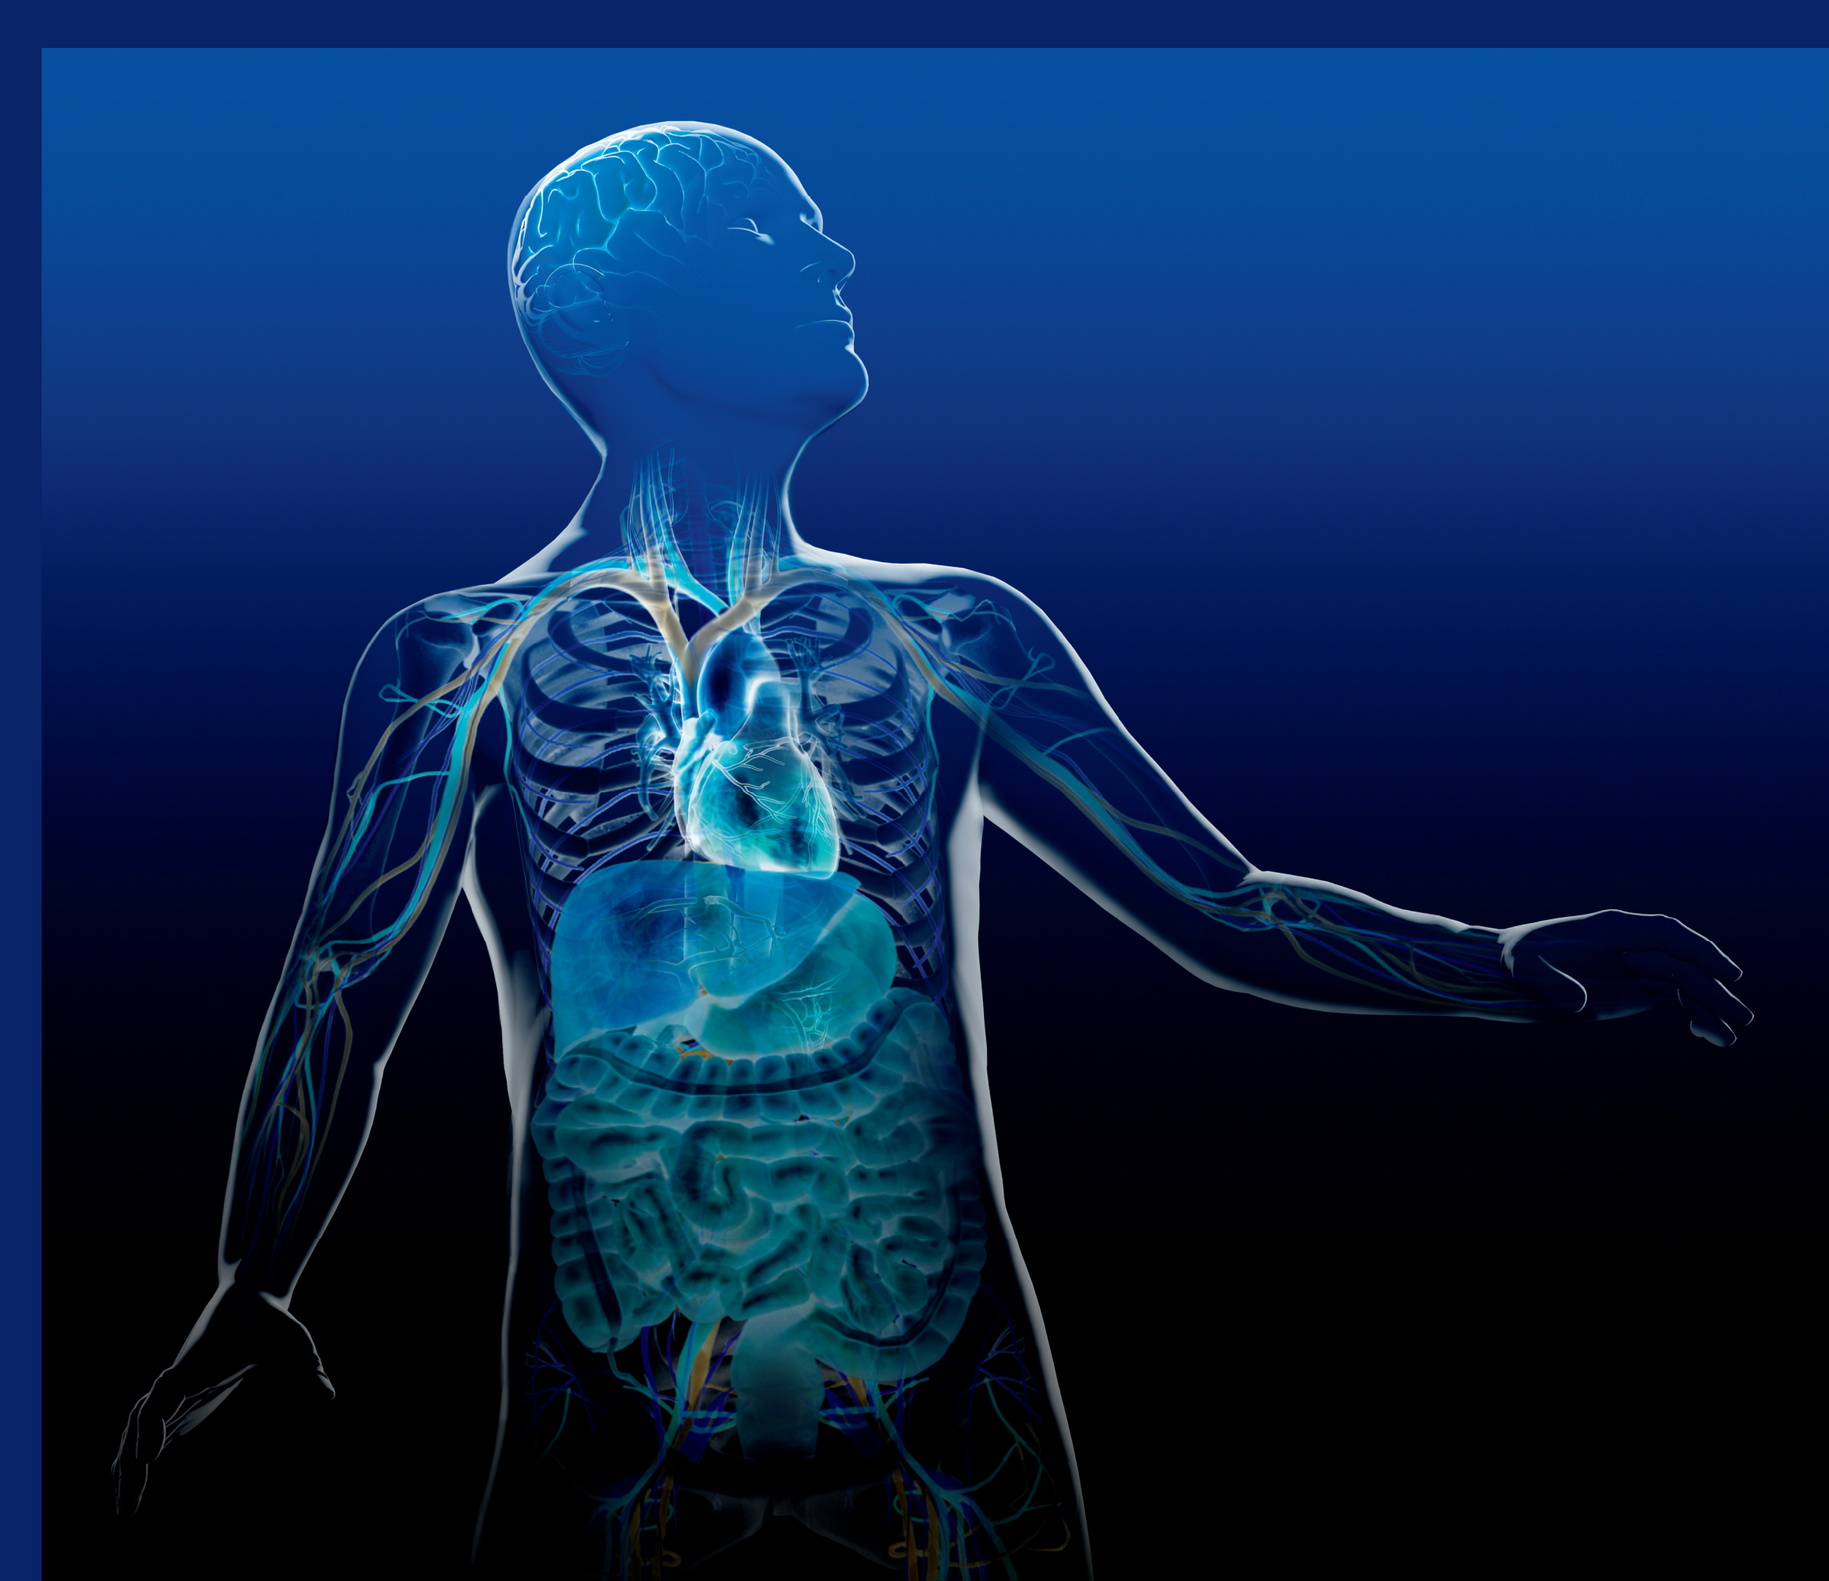

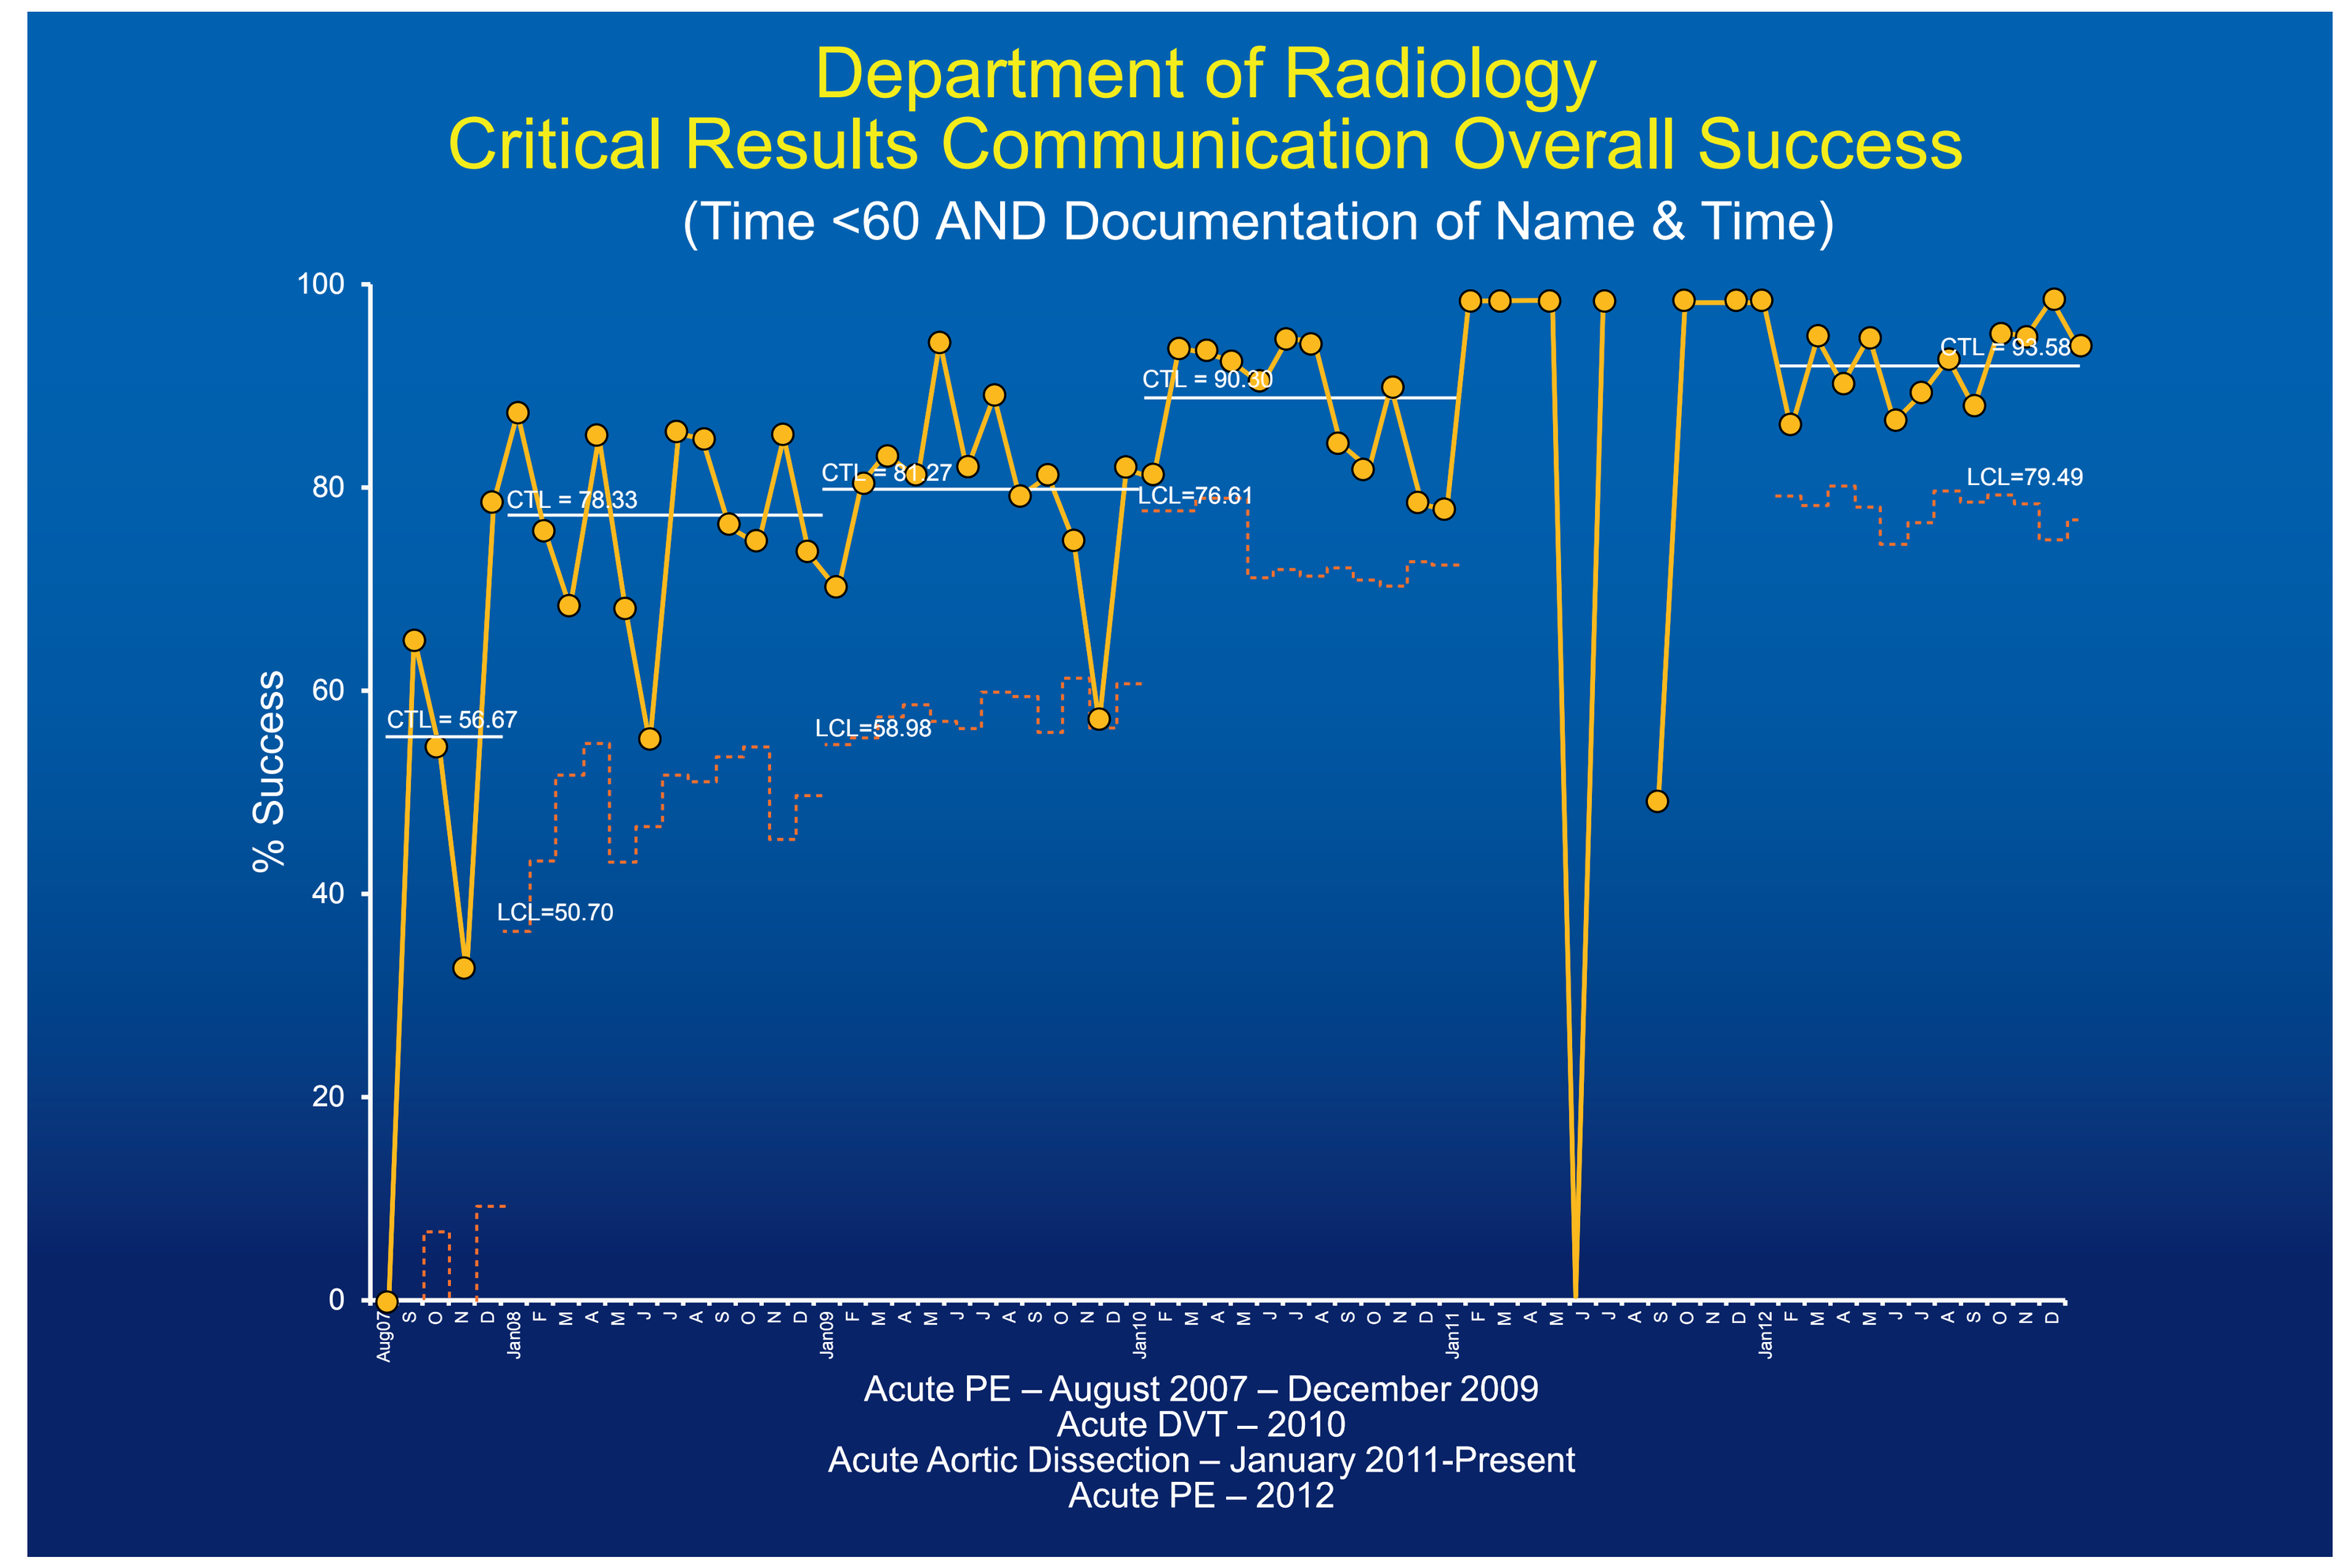
Department of Radiology
Critical Results Communication Overall Success
(Time <60 AND Documentation of Name & Time)
% Success
CTL = 56.67
CTL = 78.33
CTL = 81.27
CTL = 90.30
CTL = 93.58
LCL=50.70
LCL=58.98
LCL=76.61
LCL=79.49
Aug07
Jan08
Jan09
Jan10
Jan11
Jan12
Acute PE – August 2007 – December 2009
Acute DVT – 2010
Acute Aortic Dissection – January 2011-Present
Acute PE – 2012

Figure 2: Overall Success to Critical Results Policy
(Communicated in less than 60 minutes of recognition, documentation of licensed care provider name and time of communication in final report)

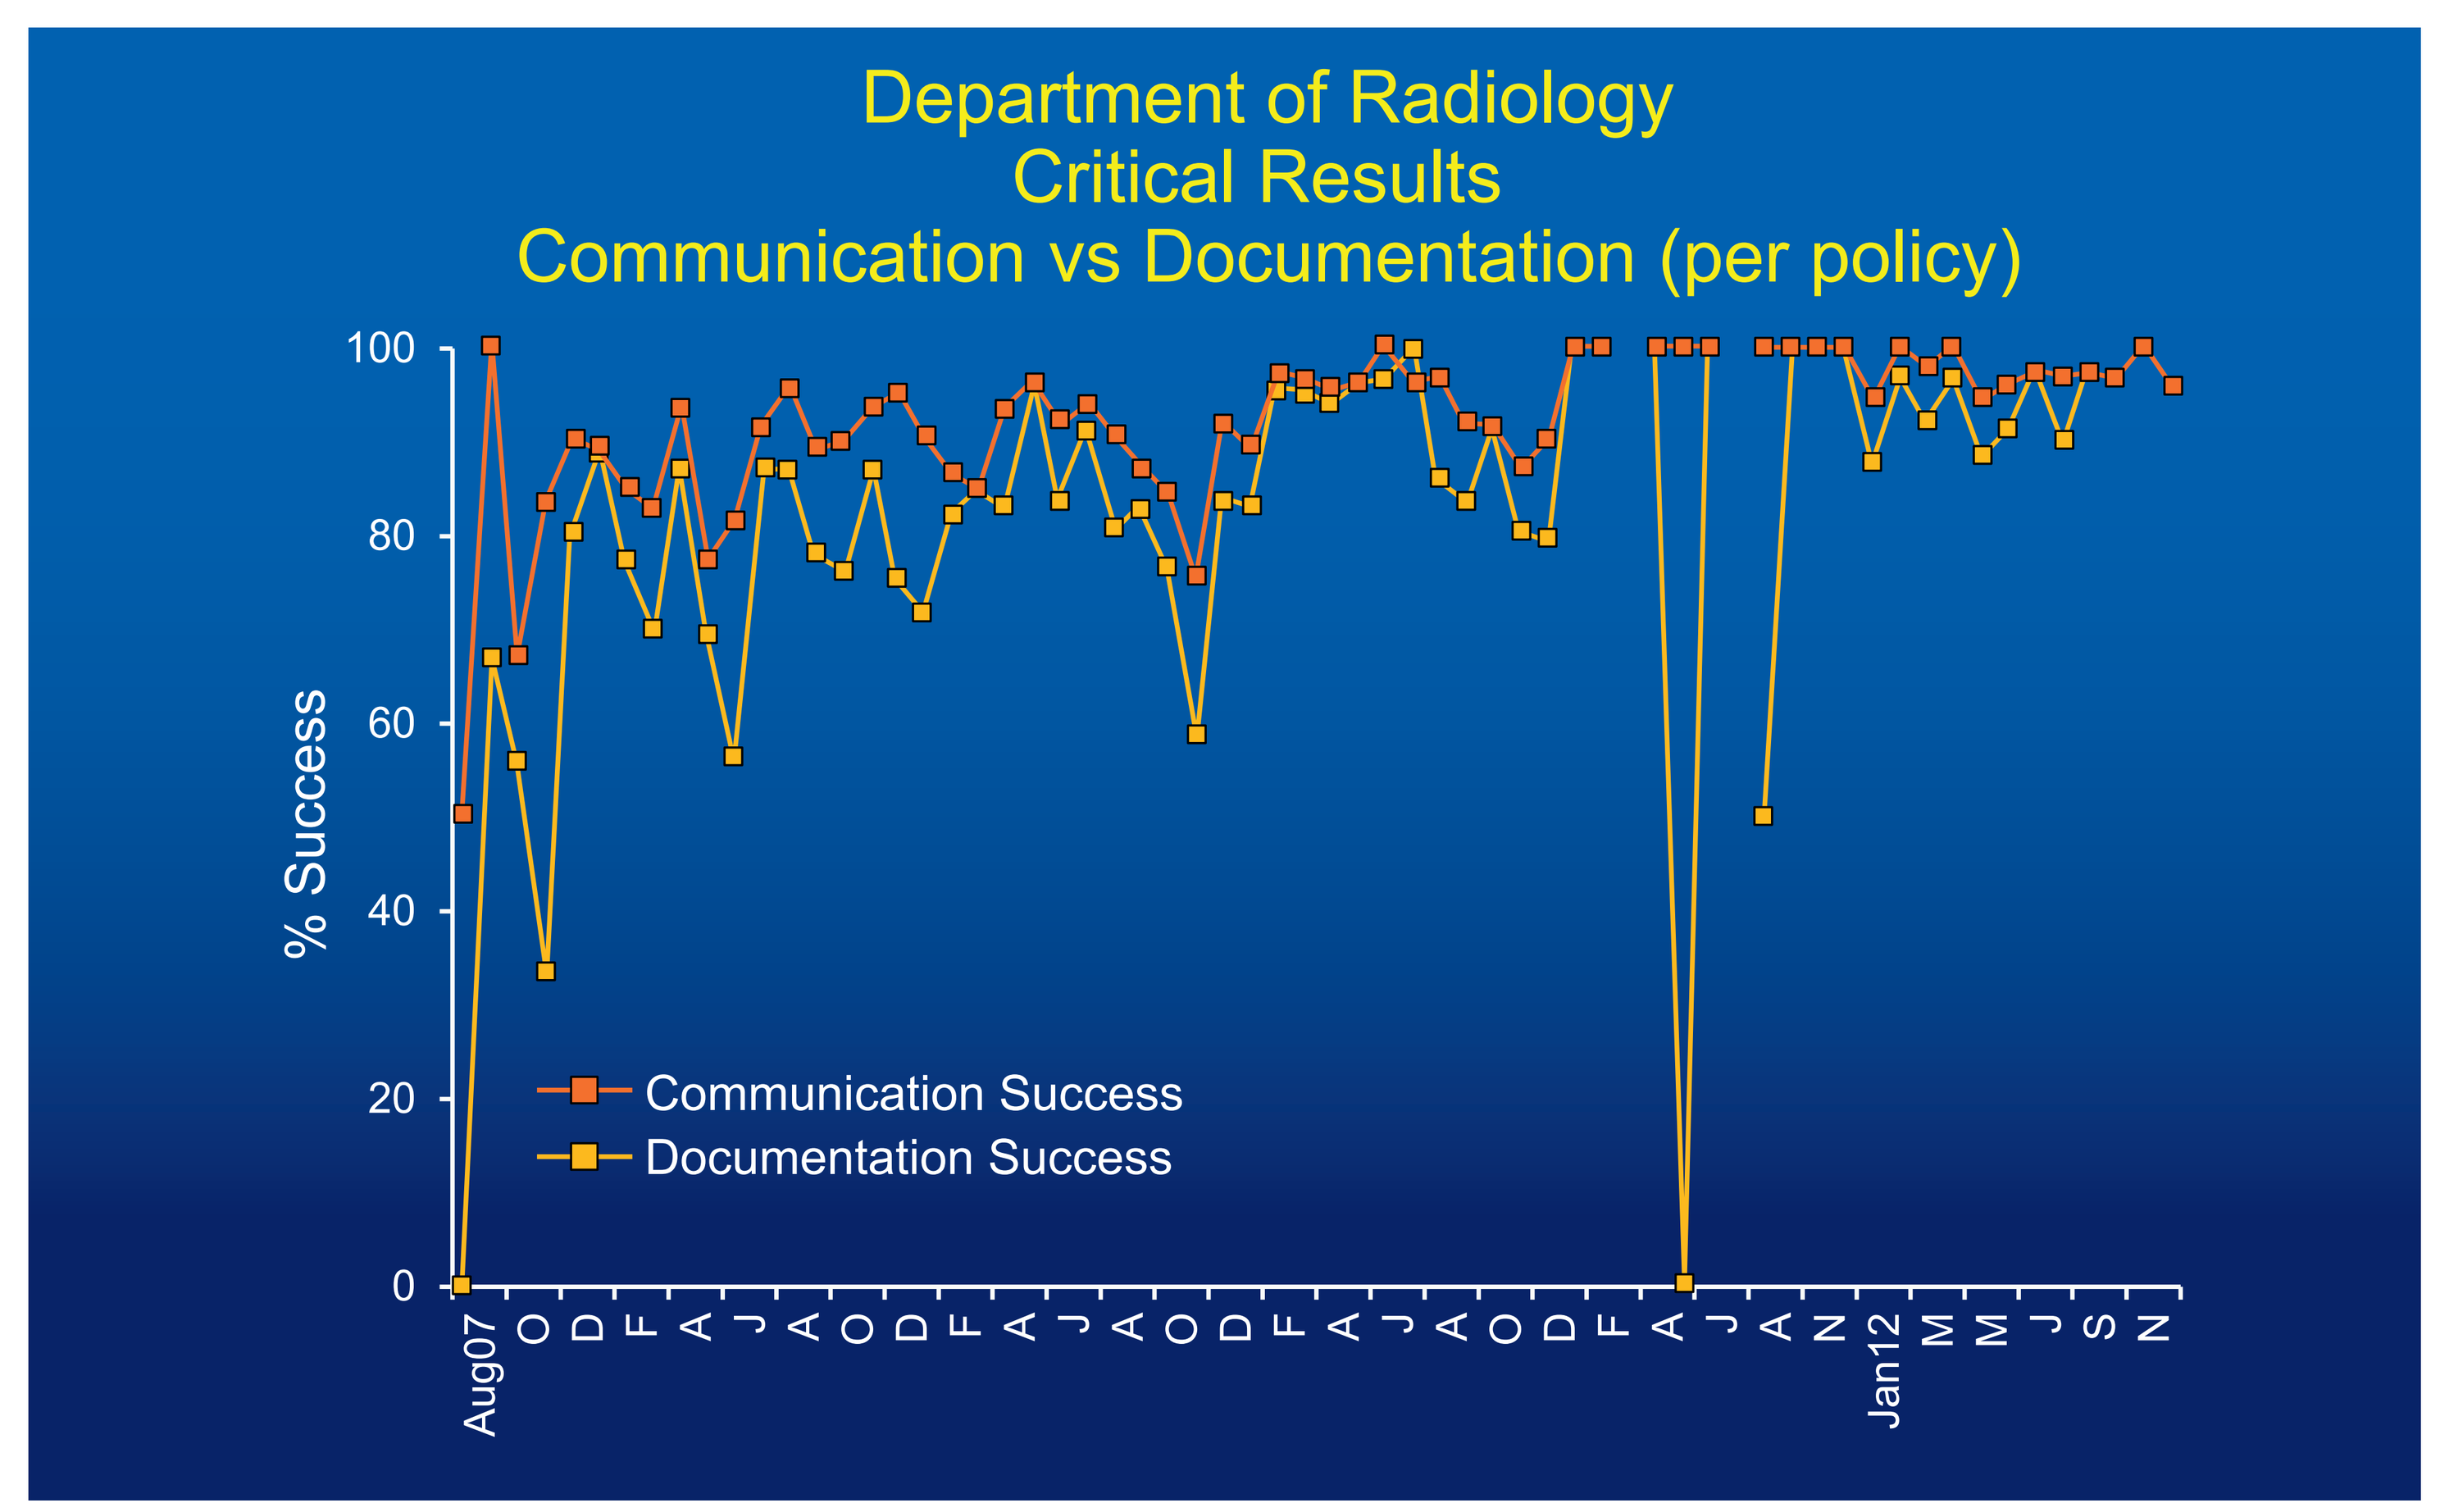
Department of Radiology
Critical Results
Communication vs Documentation (per policy)
% Success
Communication Success
Documentation Success
Aug07
Jan12

Figure 3: Difference between communicating critical finding and meeting the criteria for documentation

Improvement Efforts

1. One Critical Results Policy & Procedure for all Mayo imaging locations
2. Dedicated Institute for Healthcare (IHI) Quality Initiative focused on the barriers to effective communication of Critical Results
 - Establish process for call center to track appropriate provider when initially unsuccessful
 - Service pager required on CPOE order
 - Promote obtaining personal pager to assist in identification for report
3. Direct feedback to those radiologists who were noncompliant
4. Multiple Communication Methods
 - Emails
 - Newsletter articles
 - Presentations at division meetings and resident lectures
 - Presentations to front-line modality-specific safety champions
5. Procedural change to fast-track patients positive for acute DVT to the Thrombophilia Clinic or immediate transfer to the Emergency Department for those patients seen late in the day/after hours.

Conclusions

- A multifaceted approach to improving compliance with TJC NPSG on Critical Results requirements yielded positive, steadily improving results over the 5-year course of the study. Data revealed high compliance to timeliness of communication with opportunities for improvement related to documentation elements.
- Improved policy compliance by the radiologists assured timely care for their patients and provided effective documentation for audit and medicolegal purposes.
- Improved report mining tools would be helpful in streamlining Critical Results auditing.

© 2013 Mayo Foundation for Medical Education and Research