Improving Early Morning On-Time Start Rates at a Large Outpatient MRI Facility

Stacy R. Schultz, BA; Vicki L. Knudsen, RT(R)(MR); Jennifer L. Arndt; Jean M. Heimer, RT(R)(MR); Nicole M. Jensen, RN;
Hannah C. Kilen, RN; Patricia E. May, RT(R)(MR); Pamela K. Strike; Kirk M. Welker, MD
Department of Radiology
Mayo Clinic, Rochester, MN

Background

Mayo Clinic's Neuroradiology Outpatient MRI Center is located in the Charlton North Building and images approximately 200 patients per day. The ability to start the first patient of the day on time is critical to the following appointments throughout the remainder of the day. When this does not occur, it adds undue stress for patients and staff along with increased wait times. We describe quality improvement efforts made to increase the on-time start rate from a baseline of 17% in March 2013 to 78% in March 2014.

Define

MAYO CLINIC

Mayo Clinic Enterprise Standard Project Charter

Project Name:	**ALIVE @ 6:45 – Charlton North On-Time Start Project (2013)**		
Governance Level	3 - Department/Division	Project Size	Small
Description (255 Character Limit)	The Charlton North Building houses 12 MR scanners. Our ability to start on time impacts the efficiency of the entire day. Patients check in 30 minutes prior to their appointment and move through numerous process steps before they are ready for their exam. Patients have multiple appointments at Mayo throughout the day and their expectation is for us to be on time. A delayed MR start could make them late for future appointments. This project will examine all the process steps the patient goes through to look for ways to eliminate delays. Our goal is to improve the on-time start rate from 17% to 66% or better by December 31, 2013. (On time = patient PCIL "Exam RM" begin time @ 6:45 or earlier)		

Schedule

Start Date	08/15/2013	Finish Date	5/31/2014

Milestones

Date	Name
9/15/2013	Define
9/30/2013	Measure
10/30/13	Analyze
11/30/13	Improve
3/31/2014	Control

Business Need (SBAR)

Situation (2,000 Character Limit)	At Charlton North, 17% of the 6:45 patients are on the exam table on time and 83% are not. This project will examine all the process steps the patient goes through to look for ways to eliminate delays. We will start by creating a value stream map of all the process steps and how long each step takes.
Background (2,000 Character Limit)	Our 6:45 patients report at 6:15 and expect to start their exam on time. They move through a series of process steps and handoffs before they are ready to be brought into the exam room. Currently, only 17% of our patients are ready to start their exam by 6:45 am.

Communication Plan: ALIVE@6:45 (Charlton North On Time Start Project)

Message	Purpose / Objective	Vehicle/Media	Sender	Responsible	Audience	Frequency	Date	Status
Goals of the project Timeline Updates	**Initial Communication** (Create awareness. The objective is to ensure all stakeholders understand the project scope, high level timeline, and how the project may affect them). **Inform** (goal is to understand the current activities & overall health of the project) **Seek input** **Closure** (To ensure a smooth transition to process owners, report outcomes, reinforce the value, and recognize those involved)	How do you plan to communicate	Who will send out the communication	Who is responsible that the communication is sent	Who is the communication going to	How often	Plan for communication	When was it actually done
Introduction of Project	**Initial Communication:** Create awareness that a group has been formed to improve on time starts at Charlton North for the 6:45 appointment slots	VSM Initiative	Tech Supervisor	Tech Supervisor	Radiology Supervisors	One Time	8/20/2013	Complete
Introduction of Project	**Initial Communication:** Create awareness that a group has been formed to improve on time starts at Charlton North for the 6:45 appointment slots	MR Operations	Operations Manager	Operations Manager	MR Operations Members	One Time	9/03/2013	Complete
Introduction of Project	**Initial Communication:** Create awareness that a group has been formed to improve on time starts at Charlton North for the 6:45 appointment slots	Neuro Division	Physician Supervisor	Operations Manager	Neuro Division	One Time	9/11/2013	Complete
Introduction of Project	**Initial Communication:** Create awareness that a group has been formed to improve on time starts at Charlton North for the 6:45 appointment slots	Email	, Tech Team Member	Project Managers- Vicki/Jean	All Employees that work in MRI	One time	9/13/2013	9/12/2013

Mayo Clinic Stakeholder's Analysis Template

Stakeholders (can be Individuals / Groups / Departments)	ARCIVD Role	Key Interests & Issues	Assessment of Impact (H,M,L)	Current Status (advocate, supporter, neutral, critic, blocker)	Key Communication Points
Patients	C	Arriving on time, Sedation at Desk, Clear Communication e.g. reading the PAG, Keep the Patient informed, Eliminating redundancy, Scanned on Time, Way finding, Length of time, Waiting	H	Neutral	As Needed
Desk Personnel	R, C, I, D	MRI Safety and Patient Safety, protocols, PCIL Responsible List, RIMS Arrival Code, Check in Process, Interpreters, 6:00 AM Security	H	Advocate	At Toll Gates and any changes that affect them
Imaging Assts	R, C, I, D	No IV patients brought back, Issues taken care of, Staff on time, Patients on time, Communication Tools, Sedation	H	Advocate	At every stage/ At Toll Gates
Nursing	R, C, I, D	Start on time, Lack of Bays Open, E-protocols and Neuro Contrast Protocols, Hard Sticks, Labs, Sedation, Safety Checks	H	Advocate	At Toll Gates

(partial snapshots)

Measure

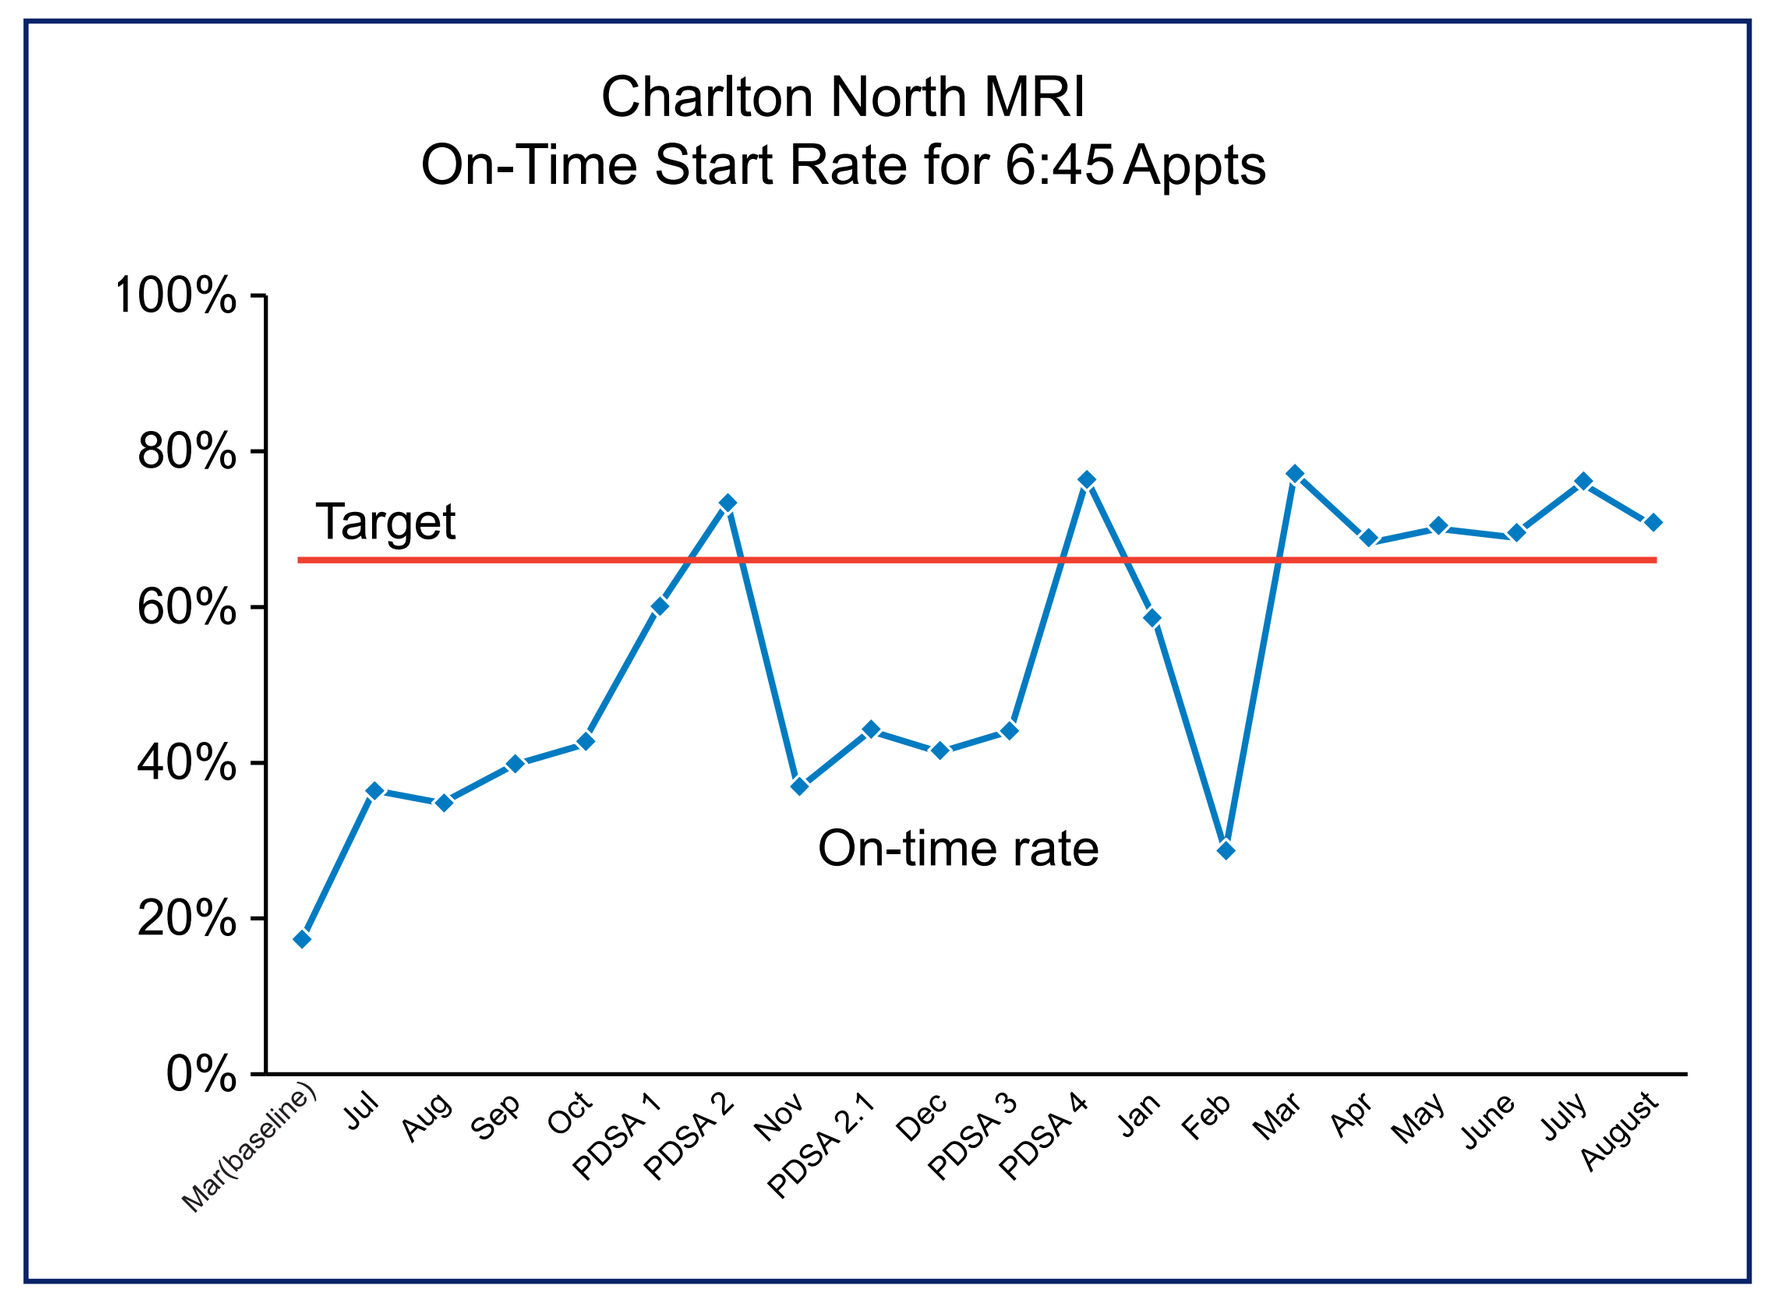

Charlton North MRI
On-Time Start Rate for 6:45 Appts
100%
80%
60%
40%
20%
0%
Target
On-time rate
Mar(baseline)
Jul
Aug
Sep
Oct
PDSA 1
PDSA 2
Nov
PDSA 2.1
Dec
PDSA 3
PDSA 4
Jan
Feb
Mar
Apr
May
June
July
August

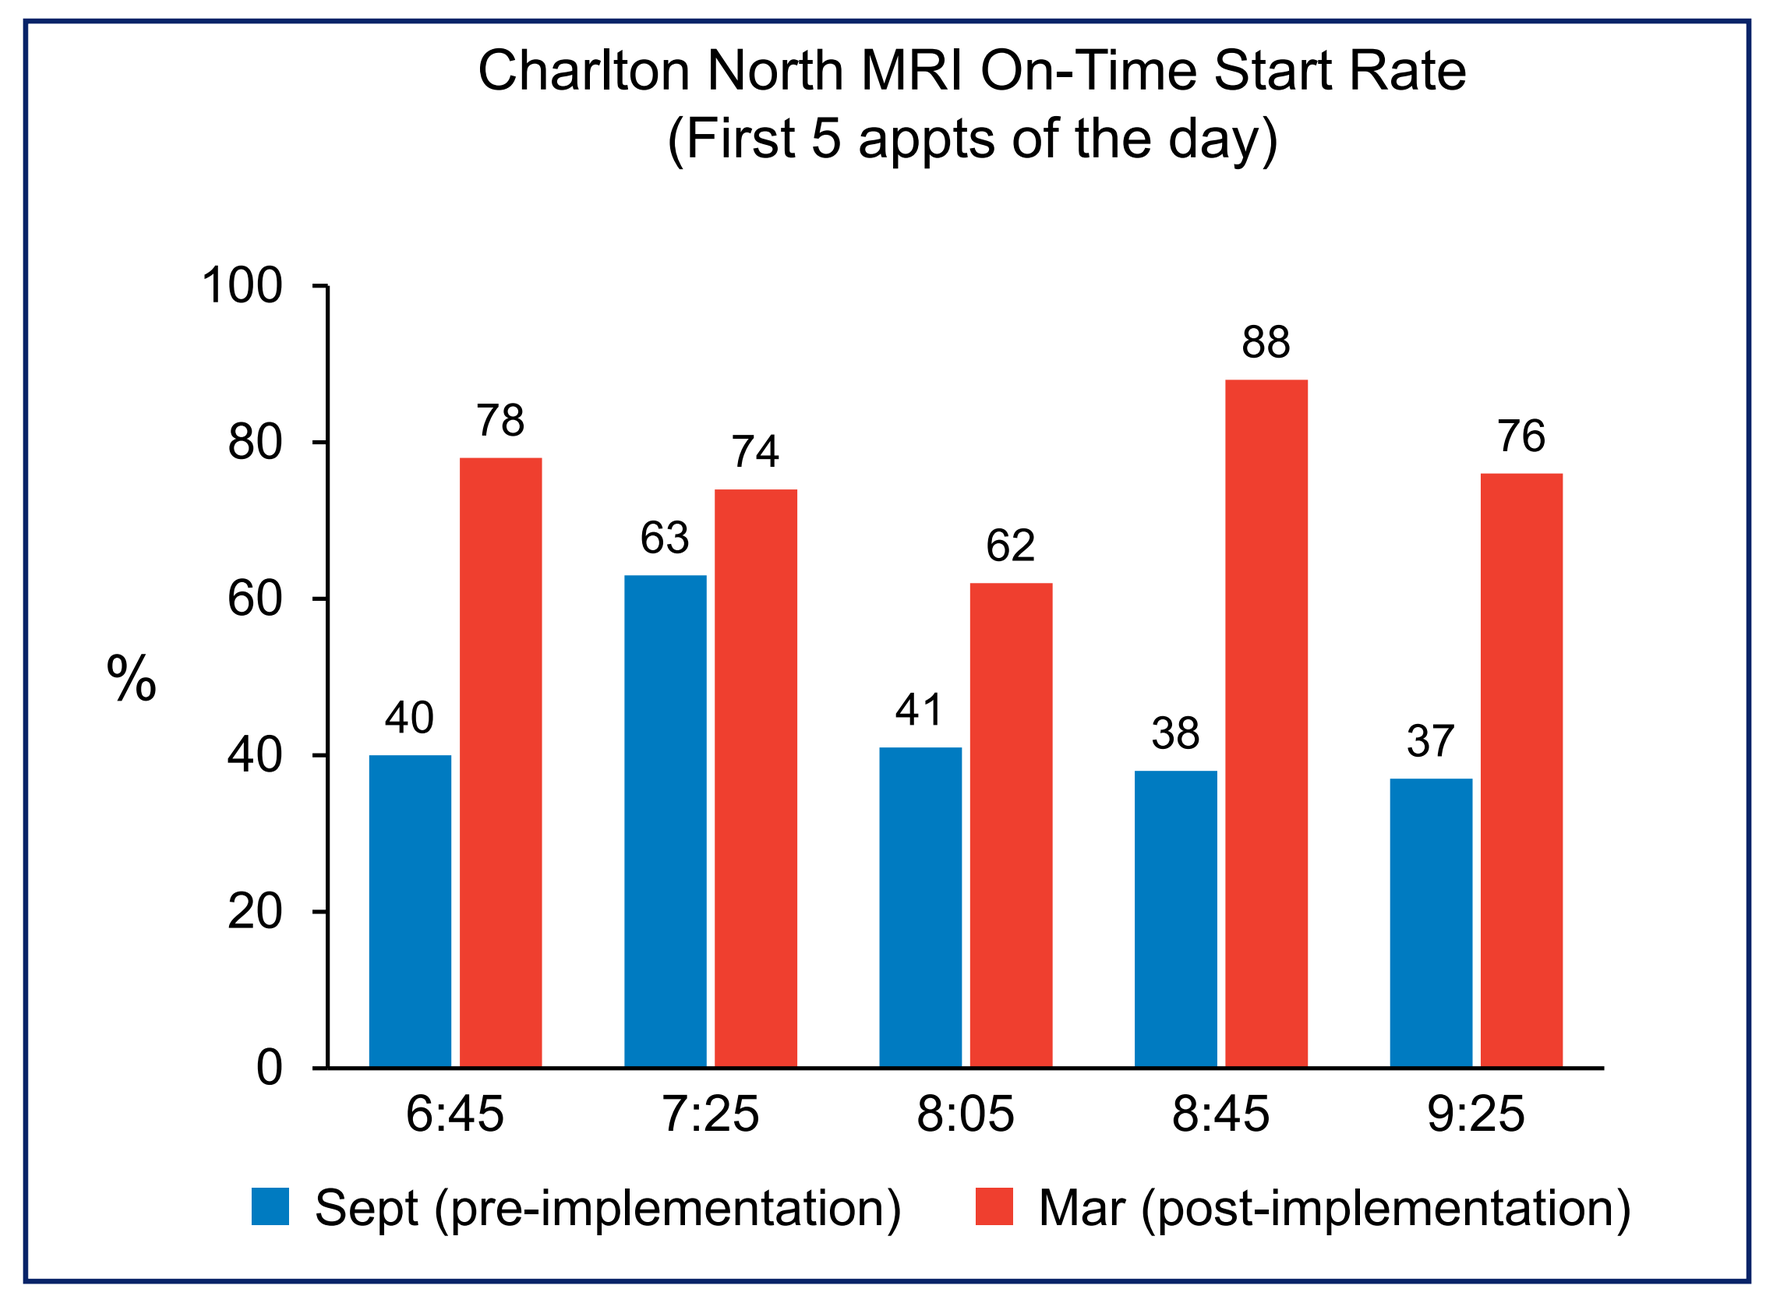

Charlton North MRI On-Time Start Rate
(First 5 appts of the day)
%
100
80
60
40
20
0
40
78
63
74
41
62
38
88
37
76
6:45
7:25
8:05
8:45
9:25
Sept (pre-implementation)
Mar (post-implementation)

Analyze

Affinity Diagram

Patient Schedule	Staff Schedule	Sedation and IV	Patient Care	Personnel	Equipment	Check In	Scoops	Facilities
No Sedation Patients before 8:05 slot **	Techs and Assts start at 6:15	No sedation patients before 8:05	6:45 patients brought right back, changed and then to assesement ***	Be ready, be ready, ready ****	Have computers up and running by start time, hence early 7 minute swipe in	No pagers at ChLn at all	Patients for next day have scoops in before leaving for the day, especially AM patients	Better directions/ education for patients new to Mayo or returning to Mayo finding ChLn
Complete Safety form prior to appt. **	2 Nurses start at 6 am	Creatinine automatically orderd at time of MRI ordered for patients greater than 69 years old ***	Make Sure patient uses restroom ***	Cross train the Tech to IA work in case of delays	Enforce no checkers until 6:45 patients are started	No pagers at ChLn at 6-7 am	Make sure PCIL list has correct check in location	
Scheduling asks weight of patient whether large bore needed.**	Hire More people **	Make sure the floor is ordering sedation before hand	tech ready for patient	Serve everyone 5 hours ene…	Alert triage if scanner …	Only 6:45 patients …		
First slots of the day have more NIV patients **	Cross train areas to help coworker when coverage needed		Patients with known ? Devices not scheduled early AM	Tec… othe… kno… wha… che…				
Peds not scheduled early AM	Schedule more RN's at 6:15 to handle 12 patients at 6:45		Nursing Instructs patient how to change into gown after starting IV	Foll… perf…				
Patient Appt start at 7:00 and tech and IA start at 6:45	Nurses assigned to a set of Bays (10,11,12)		Don't wait for patient, go find them	Ren… help… che…				
tell Patient to bring family member for sedation	Two or Three PSR's scheduled at 6:15 to get patients checked in		Have Tech bring patient to table	Put… ans… fron…				
Call Patients day before as a reminder	Start time for teams determined and consistent		IA and Techs put on Patients together, will go more quickly					
No Interpreter patients in early am slots	4 Nurses at 6:00 , 2 at 6:15			IV t… clea…				
Scheduling asks if patient has port or picc line and if sedation needed, and omit scheduling these at 6:45	IV techs, 3 at 6:00 and 1 at 6:15							
	TC start at 6:00 and prefill Bays 7,11,12,13,14,15 with IV patients and mark on PCIL sheet # of Bay							
	Another Asst in the AM							
	Flex lunch and break times around patient care time							

Key: 5 stickers; 3 stickers; 2 stickers; 1 stickers
* = number of times the idea was suggested

Impact Effort Grid

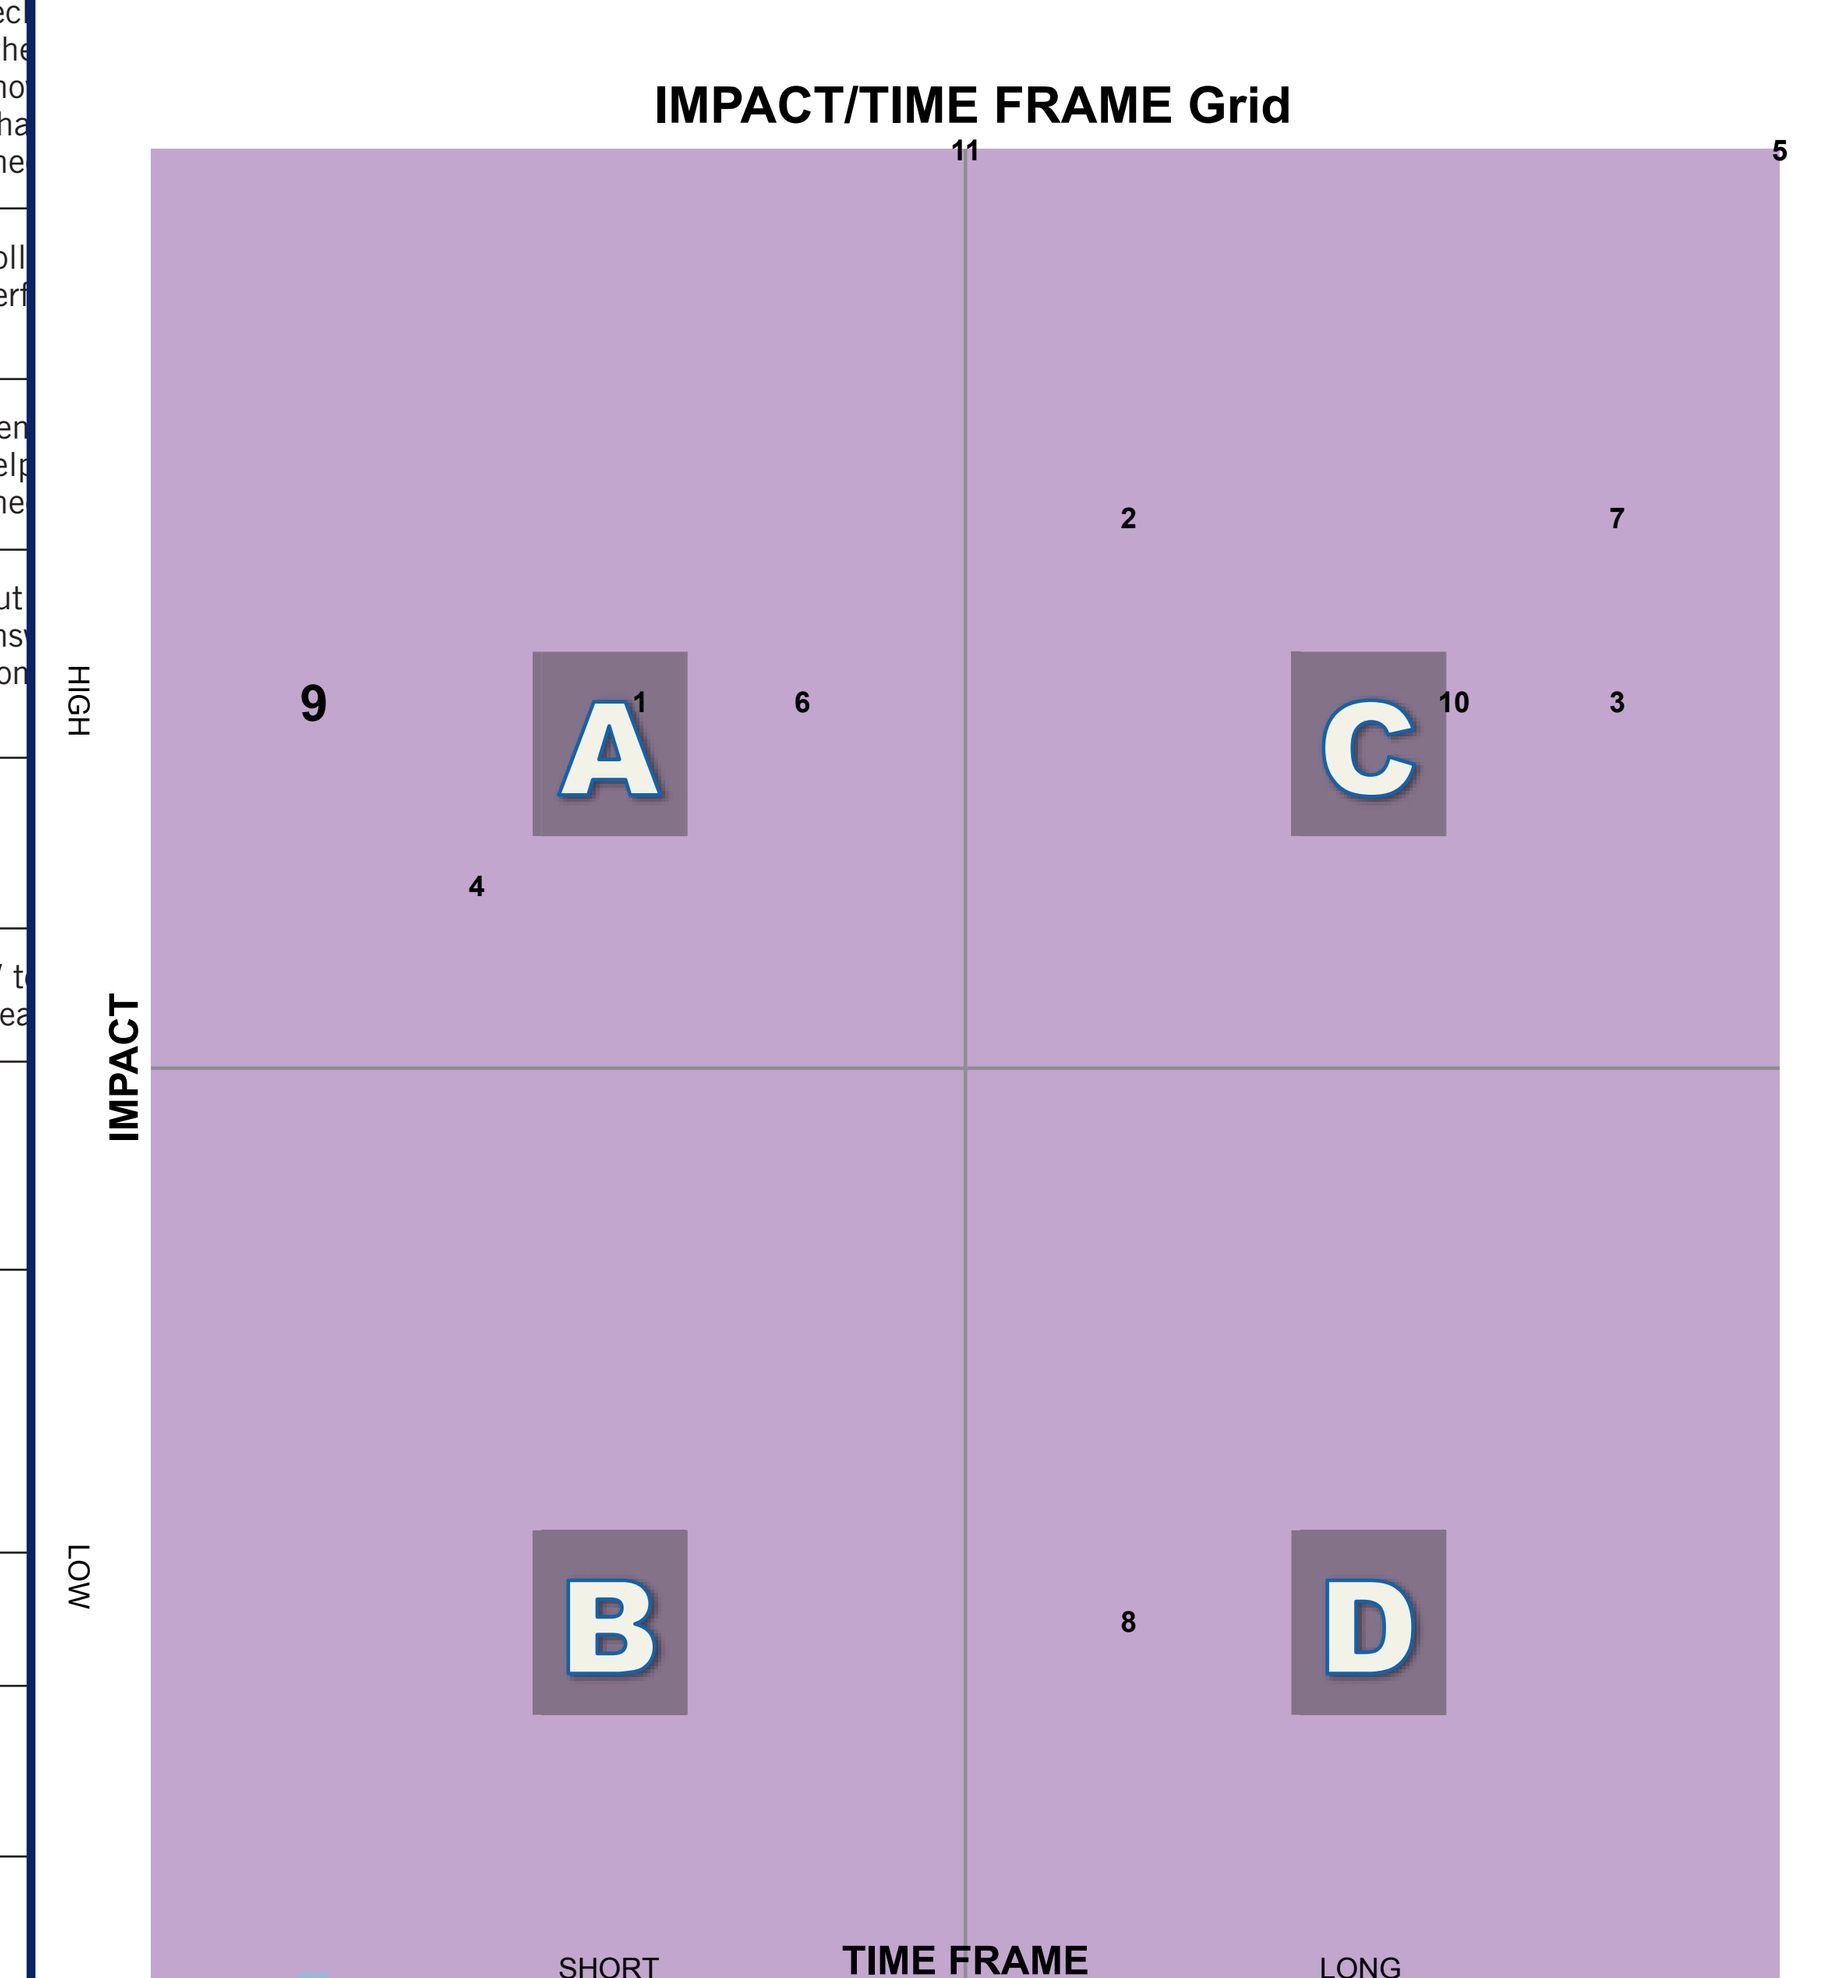

IMPACT/TIME FRAME Grid
IMPACT
HIGH
LOW
TIME FRAME
SHORT
LONG
A
B
C
D
1
2
3
4
5
6
7
8
9
10
11

(partial snapshots)

Improve

- PDSA1.0_Encouraging Restroom Usage Prior to Technologist Patient Retrieval
- PDSA2.0_Dress Patient Right Away with Imaging Assistant at 6 am
- PDSA2.1_Dress Patient Right Away with Imaging Assistant at 6 am
- PDSA3.0 Triage Tech to start at 6 am
- PDSA3.0_Imaging Assistant and Triage Tech to start at 6 am
- PDSA4.0_Staff Nurse, Imaging Assistant, & Triage Tech at 6 am
- PDSA4.1_Staff Nurse, Imaging Assistant, & Triage Tech at 6 am

Act Plan Study Do

MODEL FOR IMPROVEMENT PDSA4.0_Staff Nurse, TC, & Triage Tech at 6 am
ALIVE@6:45
Date: January 9, 2014

Objective for this PDSA Cycle: To test whether changing 6:45 am patients before assessment as well as No IV patients with a 6:00 am imaging assistant improves the 6:45 am on time start rate. The Triage tech will be starting at 6 am also to review safety sheet.

Is this cycle used to develop, test, or implement a change? To test a change

What question(s) do we want to answer on this PDSA cycle?

1. Does changing the patient before assessment show an improved on time start rate? Yes
2. Does informing the Tech "IV Started" for 6:45 patients prompt them to know that patient is changed and ready for scanner ? Yes
3. Does the lack of the MRI Safety Sheet continue to be an issue for the technologists? No
4. Is the imaging assistant better able to balance the patient load by having the IA change the No IV patients if they don't get a chance to do it? Yes
5. Does having the Triage tech there at 6 am help improve the process flow? Yes

Plan to answer questions: Who, What, When, Where

- Where: Charlton North
- What:
 - 6:45 am patients only
 - Imaging assistant, triage tech, and nurse will perform a huddle to review:
 - # of 6:45 patients for the day
 - The breakdown of IV and No IV patients
 - Review the process (lights, PCIL Stages and sedation)
 - Only the Triage tech reviews the Safety Sheet for 6:45 patients

Imaging Asst (nursing bay)

- Imaging assistant at 6:00 am to perform the following duties:
 - Do a "Print Screen" of PCIL screen showing all 6:45 patients (so they can see all 6:45 patients)
 - Retrieve the PCIL ticket (highlight 6:45 patients)
 - Sort 6:45 patients from later patients
 - Room 6:45 IV patients before NIV patients
 - Room 6:45 patients and change them (batch if possible)
 - Procedure: After the changing instructions and before imaging assistant leaves the bay.
 - Script: "Please use the restroom after you change and lock up your belongings (give directions to restroom) and when you come back, your bay number is number is XX."
 - Write Bay number on PCIL sheet and put it back in the rack for the nurse
 - Turn Bay light to white for IV and white and green for NIV
 - Move IV patients to "Subwait" if possible in PCIL, write Bay number on PCIL sheet and put in spindle for nurse
 - Move No IV patients to "Patient Ready" in PCIL

Nurse

- Procedure: Move patient in PCIL to "IV Prestart"
- Perform the assessment and do not review the safety sheet for the 6:45 patients.
- Sedate any 6:45 patient that wants sedation and is assessed without calling scanner
- Turn on bottom blue light for Triage tech to screen patient
- Turn Bay light to white and orange for IV patients
- Move IV patient to "IV Started"
- Script: "We encourage you to use the restroom at this time before you are called back for your exam (give directions to restroom and their bay number)."

Triage Tech

- Start at 6:00 am
- Responsible for all 6:45 am patients to be on time and record why they are not
- Look for blue light as trigger to when patient ready for safety screening by Technologist
- Prescreen patients prior to ready for scanner after the nurse is finished with assessment. Will screen NIV patients when they are brought back.
- If patient requires additional fact finding or relocation to a different scanner they will address this and any other issues.
- Make a notation of how many IV's, NIV's and any other information we would like to know about the 6:45 patients.
- Deselect the bottom blue light when finished

IV Tech

- Assist with cleaning bays after patient

Imaging Assistant (scanner)

- Starting at 6:30, all NIV patients still in "Waiting " will need to be brought from waiting room and into Bay by IA and changed and then moved to "Pt Ready" in PCIL

When: **Monday-Friday January 13-17, 2014**

- Communication Plan:
- On, January 10, 2014 **Jennifer A.** will contact all the assistants who work at ChLn January 13-17 at 6:45 a.m. She will inform them of the following
- On Monday through Friday, January 13-17, 2014 we are testing the imaging assistant bringing 6:45 a.m. patients to the bay starting at 6 a.m. and changing the patient before the assessment. This will include IV and No IV patients. All No IV patients still in "Waiting " at 6:30 will need to be brought back from waiting room and into Bay by IA, changed and moved to "Pt Ready" in PCIL
- On Friday January 10, **Nicole J.** will contact all the nurses who work on Monday –Friday January 13-17, 2014, to no longer review the safety sheet and that the 6:45 patient will be in the bay and changed prior to IV assessment. The nurses can move all IV patients to "IV Started" including the 6:45 patients.
- On Friday January 10, **Paddy M.** will update the techs to look for "IV Started" on 6:45 patients only as to when patient is ready for scanner. All other patients after 6:45 will be in "Patient Ready" on PCIL.
- If there are questions, the imaging assistant and nurses will contact the triage tech at 6:30 at 8-9171. **Vicki or Pat** will be on the Triage assignment during the week of Monday-Friday January 13-17, 2014.
- Stacy to pull 6:45 data on Friday, January 17, 2014

Predictions *(for questions above based on plan):*

1. Pam thinks it will continue to improve and be more efficient in the teams. On-going communication needs to continue from the workgroup and/or other communication avenues. Agree
2. Amanda thinks the morning huddle will improve the process Agree
3. Stacy thinks it will improve and be confusing at the same time. Agree
4. Jean. Paddy, Jen and Nicole smell success. It did
5. Vicki thinks it will have a positive impact on 6:45 on time starts and the techs will still want the Safety Sheet prior to bringing patient into scanning area. I think the 3 groups collaborating together will make an improvement to on time starts. It did but the safety sheet wasn't an issue this week.

Do: *Carry out the change or test; Collect data and begin analysis.*

Study: *Complete analysis of data; Compare the data to your predictions and summarize the learning*

Act: *Are we ready to make a change? Yes.*

1. *We will meet with Tim, Debbie, and Dee about changing the schedule to have TC and Triage Tech there at 6:00.*
2. *A review of the spectro phantom requirements to be reviewed by MITS and a physicist.*
3. *Triage tech to review Safety form before Nursing Assessment*
4. *Imaging assistant will fill Bay 15 on down with IV patients first*
5. *Morning huddle with the three groups improves communication and process flow*
6. *We will use this as SOP as soon as the meetings are complete with Supervisor Staff*
7. *Any unscreened 6:45 patient, Triage tech will notify scanner that it wasn't done.*

We are going to perform another test. No

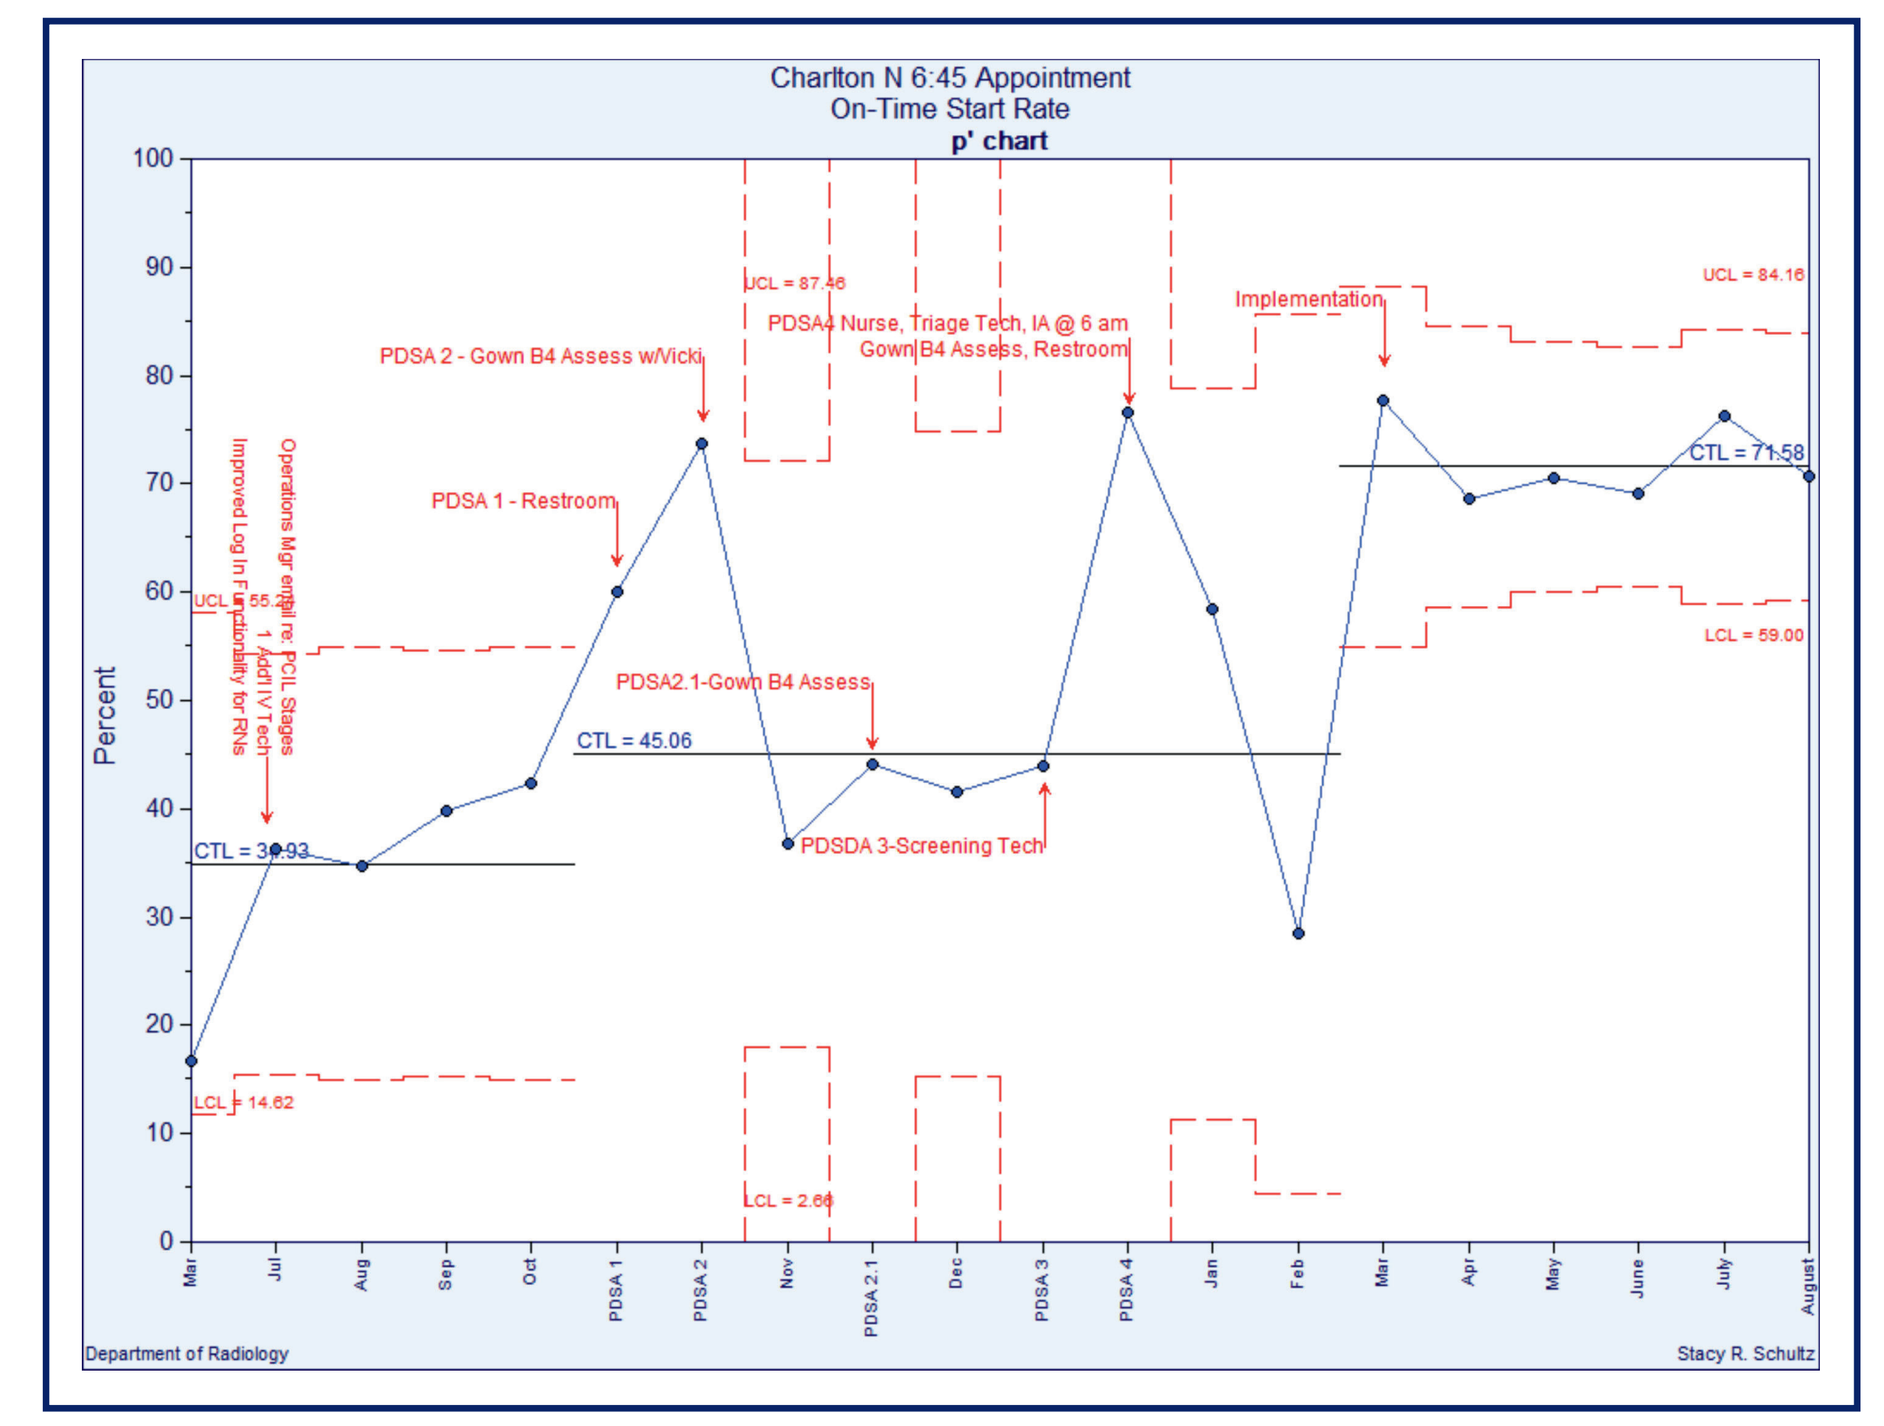

Charlton N 6:45 Appointment
On-Time Start Rate
p' chart
Percent
UCL = 87.46
PDSA4 Nurse, Triage Tech, IA @ 6 am Gown B4 Assess, Restroom
PDSA 2 - Gown B4 Assess w/Vicki
PDSA 1 - Restroom
PDSA2.1-Gown B4 Assess
PDSA 3-Screening Tech
Implementation
UCL = 84.16
CTL = 71.58
LCL = 59.00
CTL = 45.06
CTL = 34.93
LCL = 14.62
LCL = 2.66
Operations Mgr email re: PCIL Stages
Improved Log In Functionality for RNs
Department of Radiology
Stacy R. Schultz

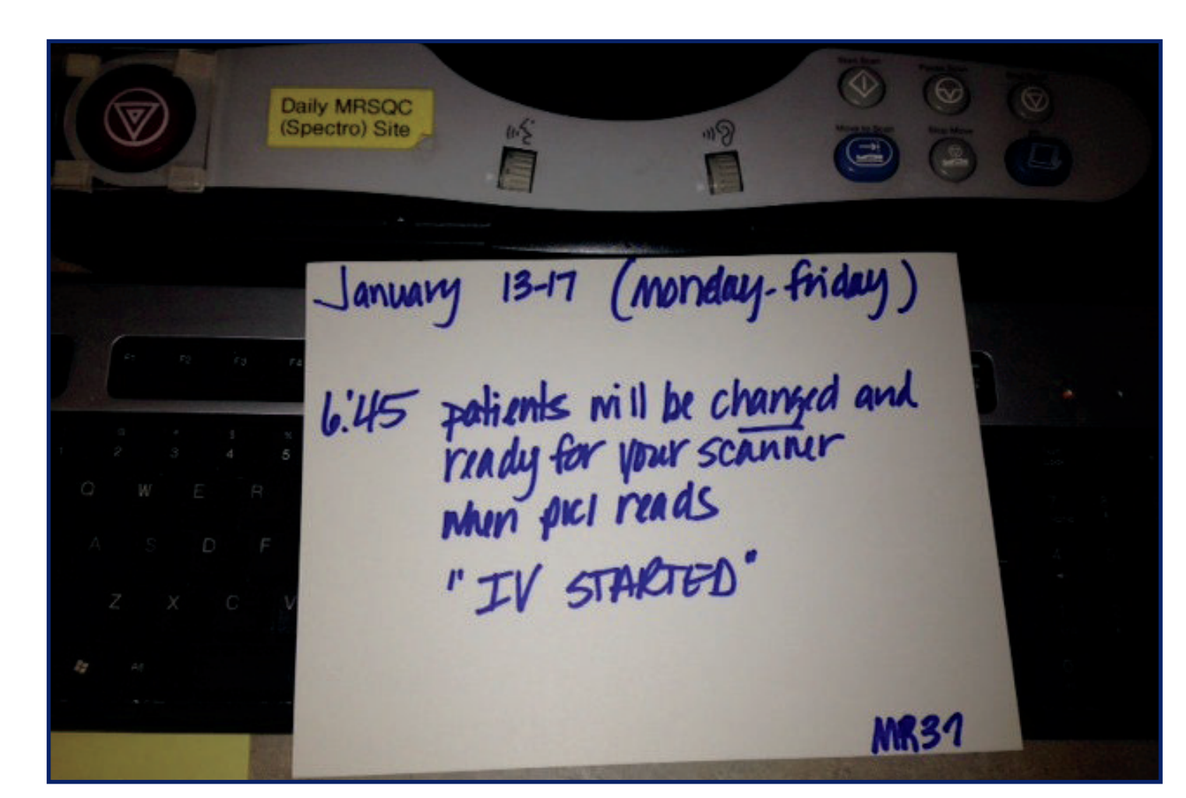

Daily MRSQC (Spectro) Site
January 13-17 (monday-friday)
6:45 patients will be changed and ready for your scanner when pcl reads "IV STARTED"

Control

- Daily monitoring for adherence to process along with monthly overall charting of on-time rate
- Continue to review staffing models for early morning coverage
- Continuous communication to all staff with regards to process steps

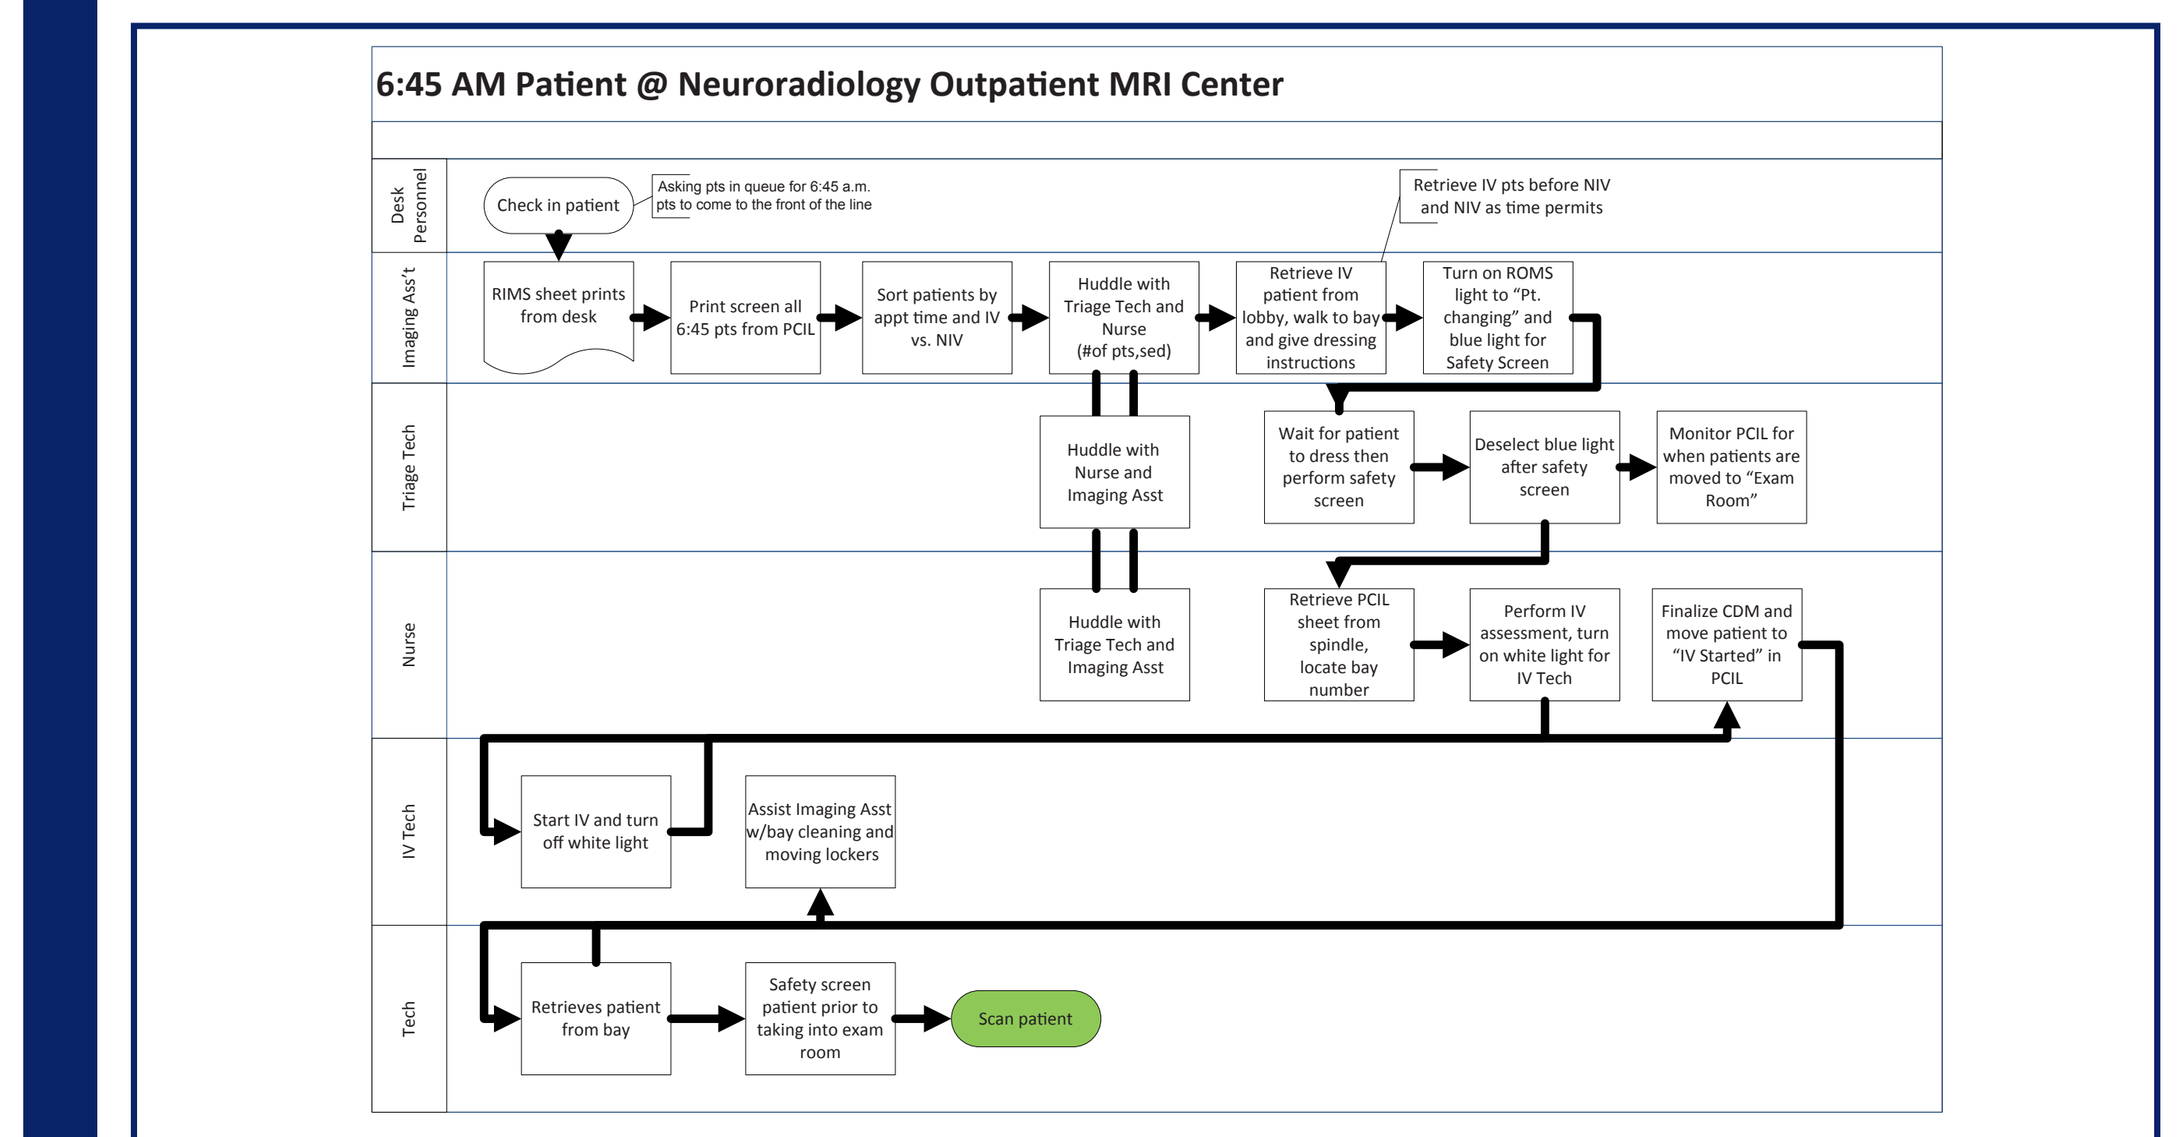

6:45 AM Patient @ Neuroradiology Outpatient MRI Center
Desk Personnel
Imaging Ass't
Triage Tech
Nurse
IV Tech
Tech
Check in patient
Asking pts in queue for 6:45 a.m. pts to come to the front of the line
Retrieve IV pts before NIV and NIV as time permits
RIMS sheet prints from desk
Print screen all 6:45 pts from PCIL
Sort patients by appt time and IV vs. NIV
Huddle with Triage Tech and Nurse (#of pts,sed)
Retrieve IV patient from lobby, walk to bay and give dressing instructions
Turn on ROMS light to "Pt. changing" and blue light for Safety Screen
Huddle with Nurse and Imaging Asst
Wait for patient to dress then perform safety screen
Deselect blue light after safety screen
Monitor PCIL for when patients are moved to "Exam Room"
Huddle with Triage Tech and Imaging Asst
Retrieve PCIL sheet from spindle, locate bay number
Perform IV assessment, turn on white light for IV Tech
Finalize CDM and move patient to "IV Started" in PCIL
Start IV and turn off white light
Assist Imaging Asst w/bay cleaning and moving lockers
Retrieves patient from bay
Safety screen patient prior to taking into exam room
Scan patient

Project Closure Report

Project Name: ALIVE@6:45 Project
Departments: Emergency and Radiology
Product/Process: First patient on scanner by 6:45

TABLE OF CONTENTS

1 PROJECT CLOSURE REPORT PURPOSE 4
2 PROJECT CLOSURE REPORT GOALS 4
3 PROJECT CLOSURE REPORT SUMMARY 4
3.1 Project Background Overview 4
3.2 Governance and Project Organizational Structure 5
3.3 Project Highlights and Best Practices (That Which We Want to Continue) 5
3.4 Project Top Five Lessons Learned 5
3.5 Project Closure Synopsis 5
4 SCOPE VERIFICATION 6
4.1 Goals and Objectives Performance 6
4.2 Success Criteria Performance 6
4.3 Milestone and Deliverables Performance 6
4.4 Quality Management 6
5 PROJECT METRICS PERFORMANCE 7
5.1 Schedule Performance 7
5.2 Budget Performance 7
5.3 Issue Management 7
5.4 Risk Management 7
5.5 Communication Management 8
5.6 Metrics Performance Recommendations 8
6 OPERATIONS TRANSITION PLAN 8
6.1 Organizational Structure 8
6.2 Resource Management 8
6.3 Customer Expectation Management 8
6.4 Asset Management 9
7 PROJECT CLOSURE TASKS 9
7.1 Postproject Tasks 9
7.2 Project Closure Recommendations 9
8 PROJECT CLOSURE REPORT APPROVALS 10
9 APPENDICES 11
9.1 Project Closure Checklist Documents 11

Lessons Learned

- Huddles and ongoing communication were key to teamwork
- Process observations at the beginning would have been helpful
- Debunking the myths as to the "perceived" root causes
- Staffing model was not conducive to accommodate 6:45 am patients
- Working differently for staff was difficult at first but made a huge difference once all were "on board"
- Redundancy slows down the process
- Focus on the processes and not the people
- PDSA cycles conducted during the holidays were difficult

For questions, scan here to contact us.

© 2014 Mayo Foundation for Medical Education and Research