Clinically-based Imaging Algorithm for More Efficient Imaging Utilization in Suspected Appendicitis in a Pediatric Population

Ankaj Khosla, MD[1]; Li Ern Chen, MD[2]; Mohammed Badawy, MD[3]; Rodica Pop, PhD[4]; Jeannie Kwon MD[1]

1. Department of Radiology, University of Texas Southwestern Medical Center, Children's Medical Center Dallas
2. Department of Surgery, Baylor University Medical Center Dallas

Background

Acute appendicitis is the most common abdominal condition requiring surgery in children. At our tertiary care children's hospital, we treat more than 1100 children with appendicitis yearly. Diagnostic imaging, namely, ultrasound and CT, is an integral part of the workup for suspected appendicitis, with variable utilization practices dependent upon the clinical provider. Therefore, we developed an algorithm to aid in more efficient imaging modality selection for pediatric patients suspected of having appendicitis.

Aims

To develop a clinically based imaging algorithm for more efficient radiology utilization in patients suspected of having appendicitis and reducing ionizing radiation exposure.

Methods

- A multidisciplinary team from the emergency room, surgery and radiology departments convened to develop diagnostic imaging algorithm (Figure 1) for patients with suspected appendicitis.
- Four deficiencies identified: indications for diagnostic imaging, appropriate study choice (US vs. CT), technical performance in imaging, and consistency in radiologic reporting.
- Emergency room physicians begin using pediatric appendicitis score (PAS)[1], assigning patients to a low, moderate or high-risk based on history, clinical and laboratory findings.
- Imaging studies only used in a moderate-likelihood of appendicitis; ultrasound in patients with BMI < 30 and CT in patients with BMI ≥ 30 or suspected abscess.
- Improved sonographic study performance with a standardized protocol on conducting exams for appendicitis with technologists receiving didactic and hands-on training on sonographic techniques and adequate pain control provided by emergency room physicians.
- Radiologists provided standard reporting template with specific categorizations for making a diagnosis.
- Data was gathered before and after implementation of the imaging pathway to track the imaging studies being ordered, as well as the rate of missed appendicitis. The pre-pathway group consisted of all patients with appendicitis from January 2011 to December 2011. An interval of nine months was provided to implement the algorithm and train all providers at the hospital. Post-pathway data was gathered from October 2012 to December 2013. Data continued to be gathered and a validation group was created of patients from January 2014 to June 2014.
- Fisher's test was performed to compare the pre-pathway and post-pathway and validation groups.

Results and Conclusions

- By implementing a multidisciplinary approach, we developed an imaging algorithm for the work-up of suspected appendicitis in a pediatric population.
- Algorithm has resulted in improvement of the missed diagnosis rate while decreasing CT utilization. (Table 1, Figures 3 - 8)
- We utilized a standardized departmental protocol for performing sonographic studies for appendicitis and a template for reporting the results in standardized fashion. (Figure 2).
- This algorithm can be implemented at other institutions and validated to determine its efficacy.

Next Steps:
We plan on further validating our algorithm by continuing to monitor outcomes and sharing our data with the pediatric, radiology and emergency medicine communities nationwide to see if they get similar results.
In addition, we plan on implementing further improvements to bring our missed appendicitis rate to as low as possible.

Figure 1: Appendicitis Clinical Care Flow Chart (algorithm)

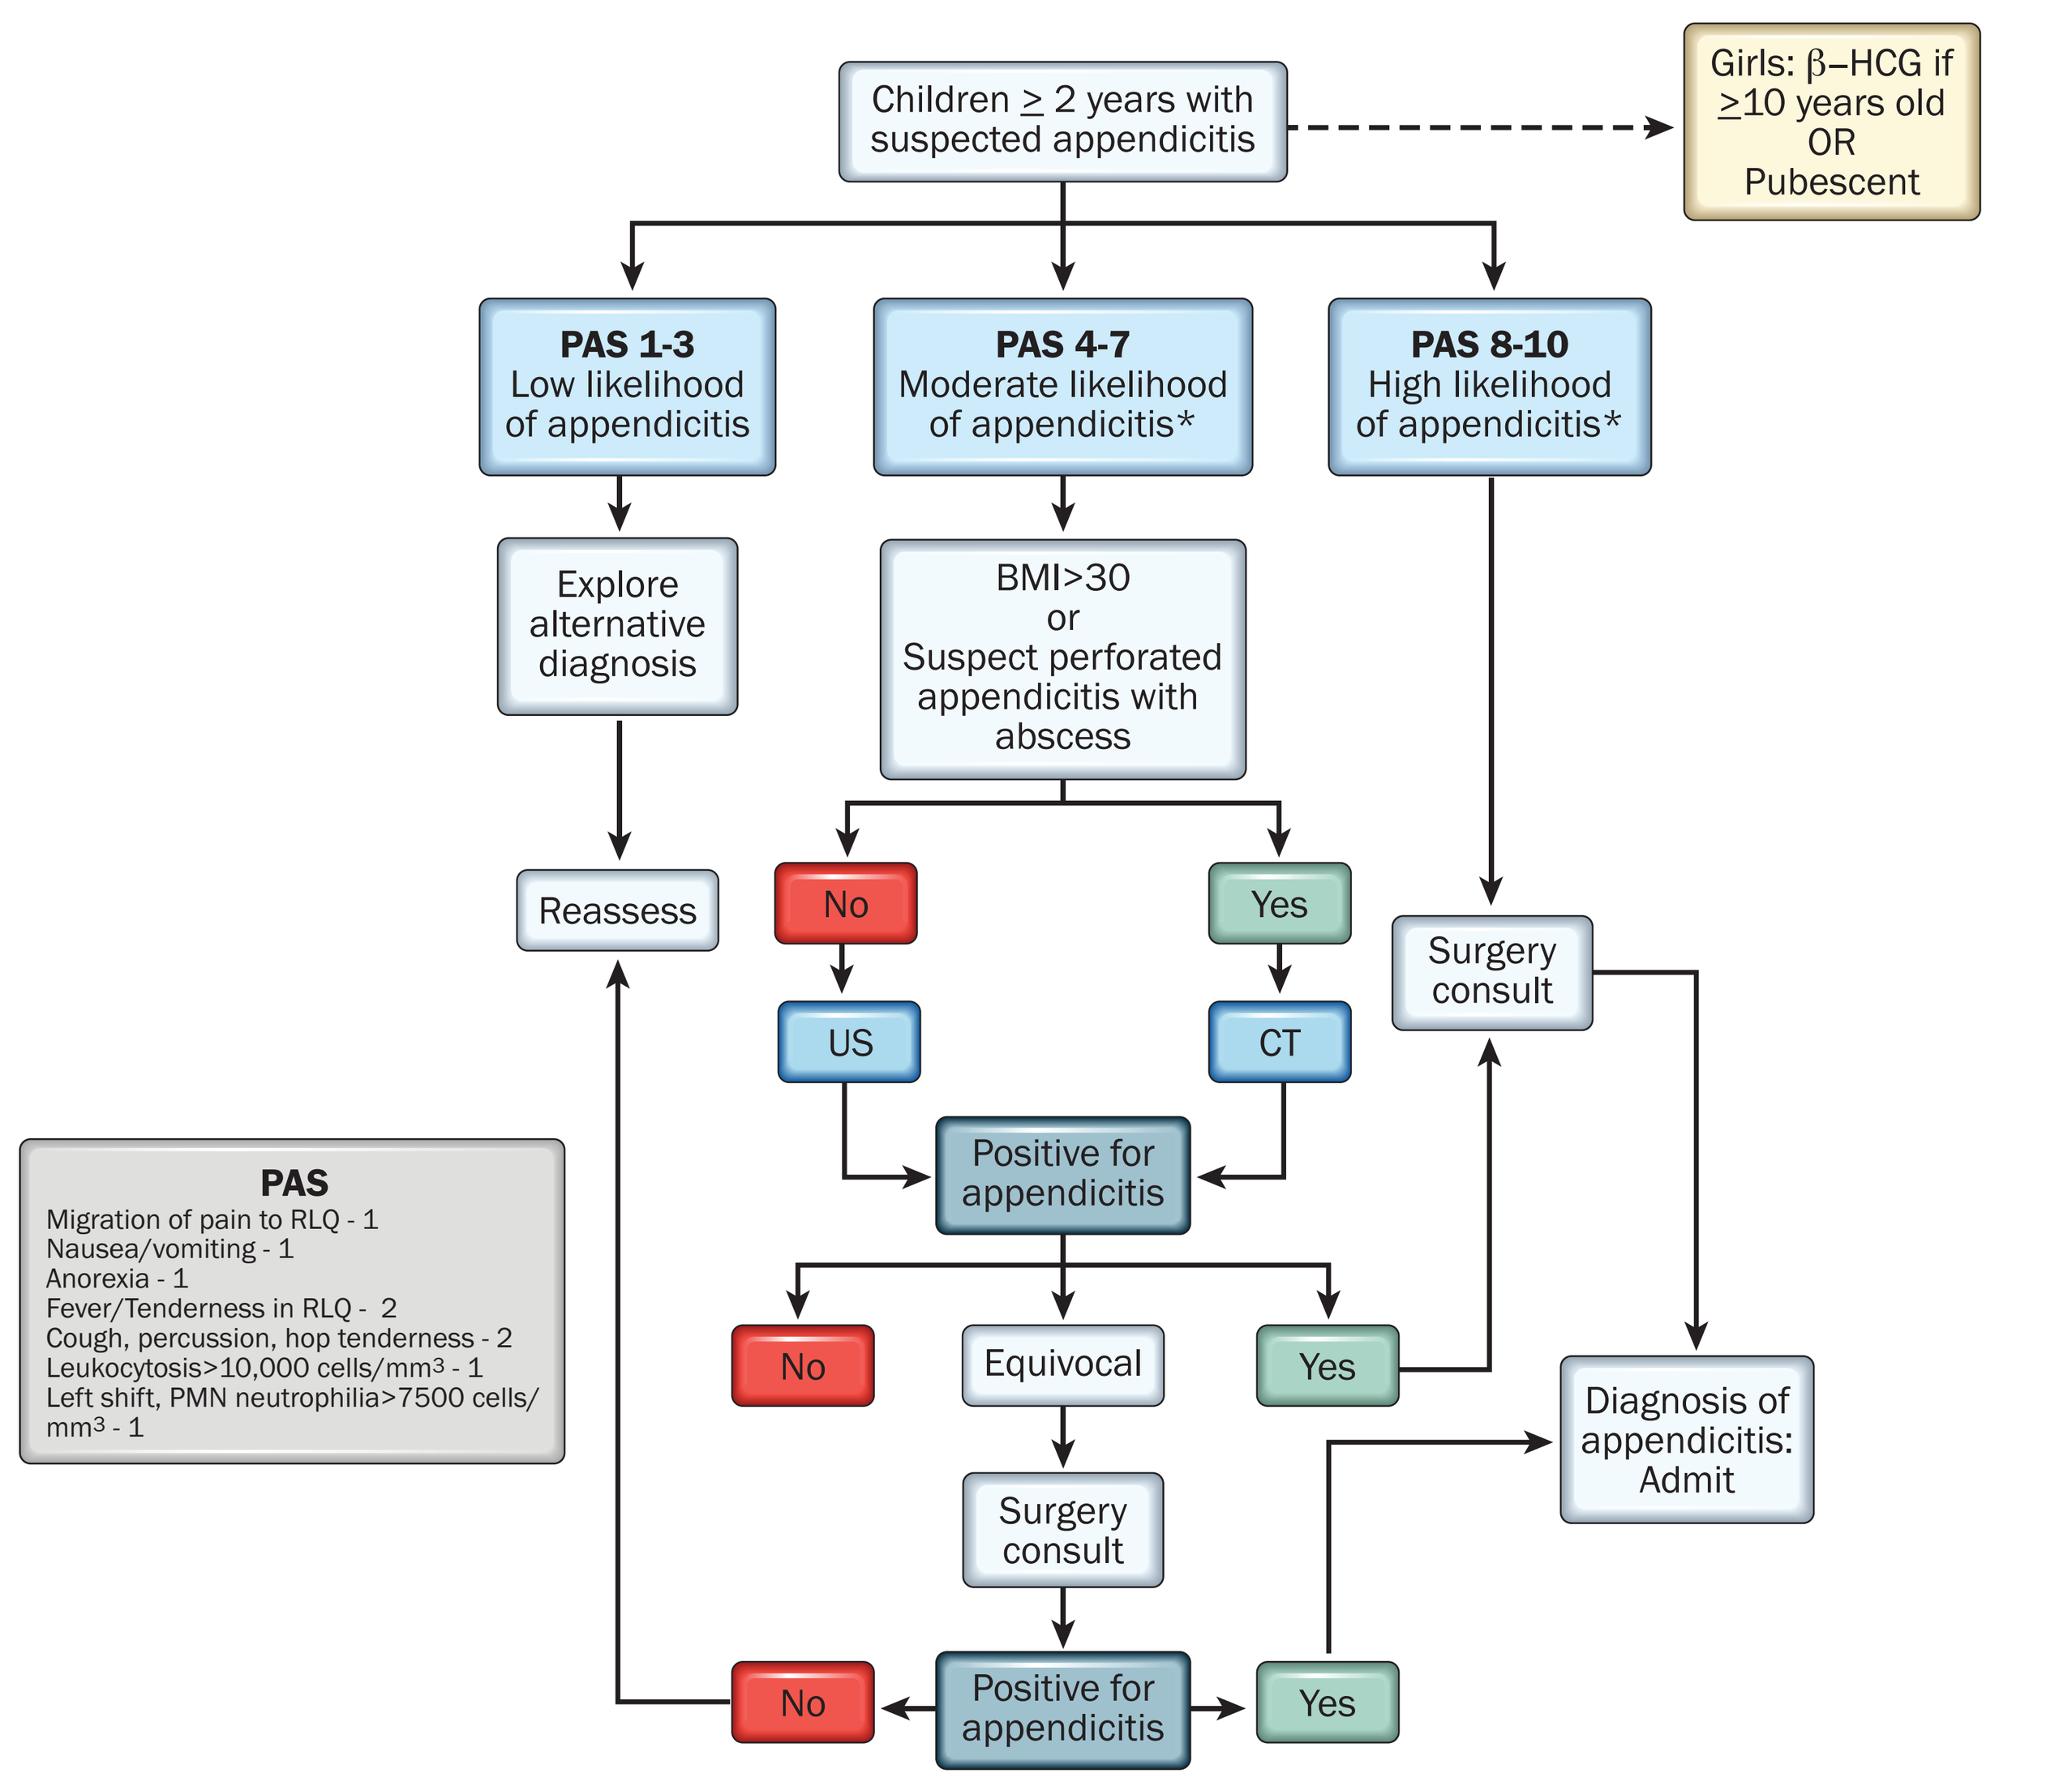

Children ≥ 2 years with suspected appendicitis
Girls: β–HCG if ≥10 years old OR Pubescent
PAS 1-3 Low likelihood of appendicitis
PAS 4-7 Moderate likelihood of appendicitis*
PAS 8-10 High likelihood of appendicitis*
Explore alternative diagnosis
BMI>30 or Suspect perforated appendicitis with abscess
Reassess
No
Yes
US
CT
Surgery consult
Positive for appendicitis
No
Equivocal
Yes
Diagnosis of appendicitis: Admit
Surgery consult
No
Positive for appendicitis
Yes
PAS
Migration of pain to RLQ - 1
Nausea/vomiting - 1
Anorexia - 1
Fever/Tenderness in RLQ - 2
Cough, percussion, hop tenderness - 2
Leukocytosis>10,000 cells/mm3 - 1
Left shift, PMN neutrophilia>7500 cells/mm3 - 1

Figure 2: Standard Report Template for Appendix Ultrasound

TECHNIQUE:
Sonographic evaluation of the right lower quadrant was performed with graded compression to evaluate for acute appendicitis. Grayscale and color Doppleer images were obtained. Survey images of the left lower quadrant, inferior right upper quadrant, periumbilical area, and midline pelvis were also obtained.

FINDINGS:
Primary findings
Appendix seen: [Yes/No/partial, location]
Maximum transverse outer diameter: [<n/a> mm]
Compressibiility: [Yes/No/ n/a]
Hyperemia: [Yes/No/ n/a]
Fecalith/appendicolith: [Yes/No/ n/a]

[Other findings or comments]

Secondary findings
Free fluid: [Yes/No/amount/location]
Fluid collection: [Yes/No/size/location]
Echogenic fat/inflammatory changes: [Yes/No]

IMPRESSION: (choose one)
[Acute appendicitis]
[Equivocal for acute appendicitis]
[Negative for appendicitis; normal appendix]

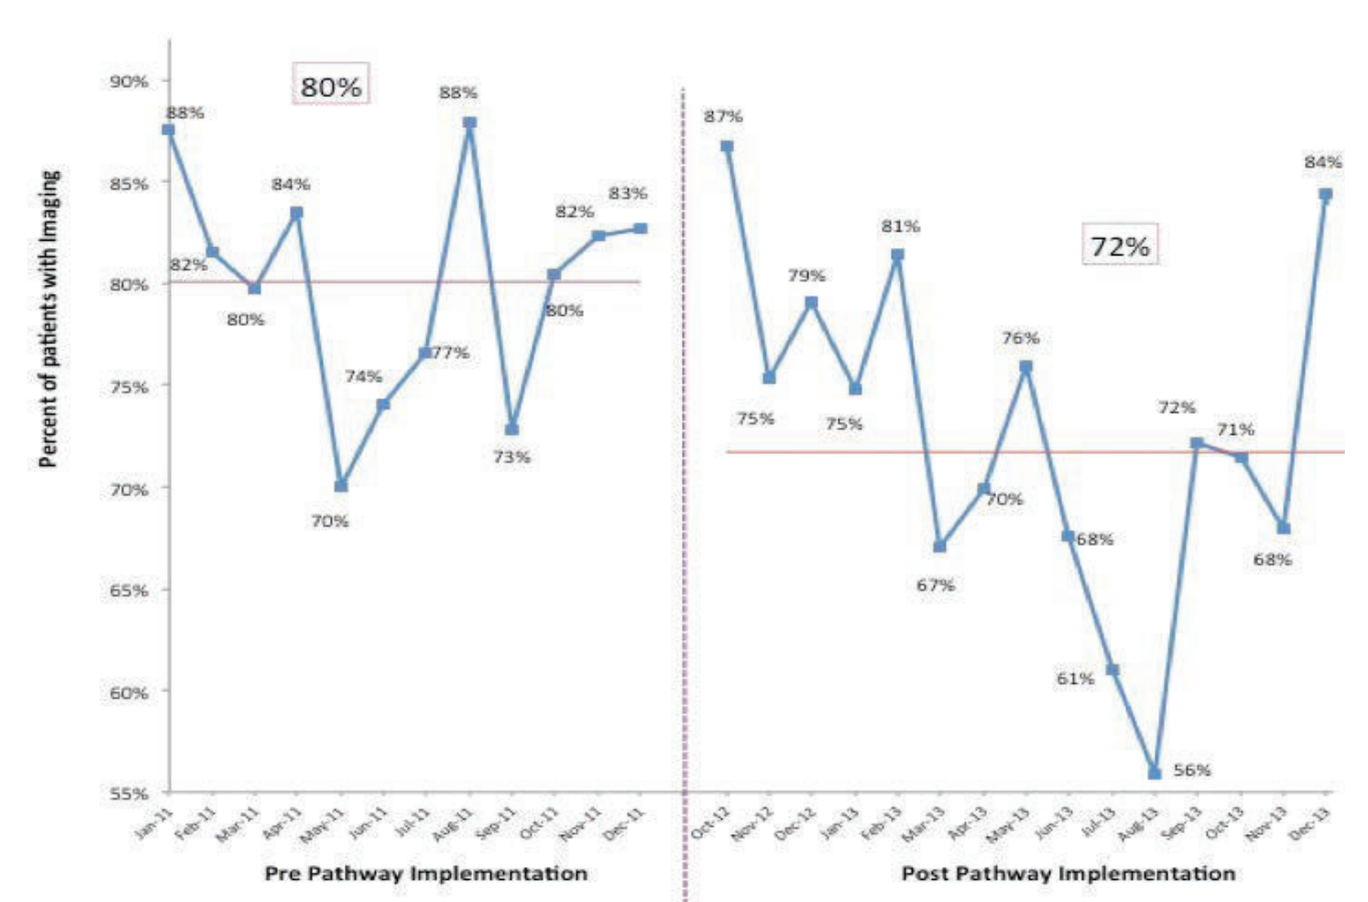

80%
72%
Percent of patients with imaging
Pre Pathway Implementation
Post Pathway Implementation

Figure 3.
Overall imaging utilization between pre- and post-pathway implementation.

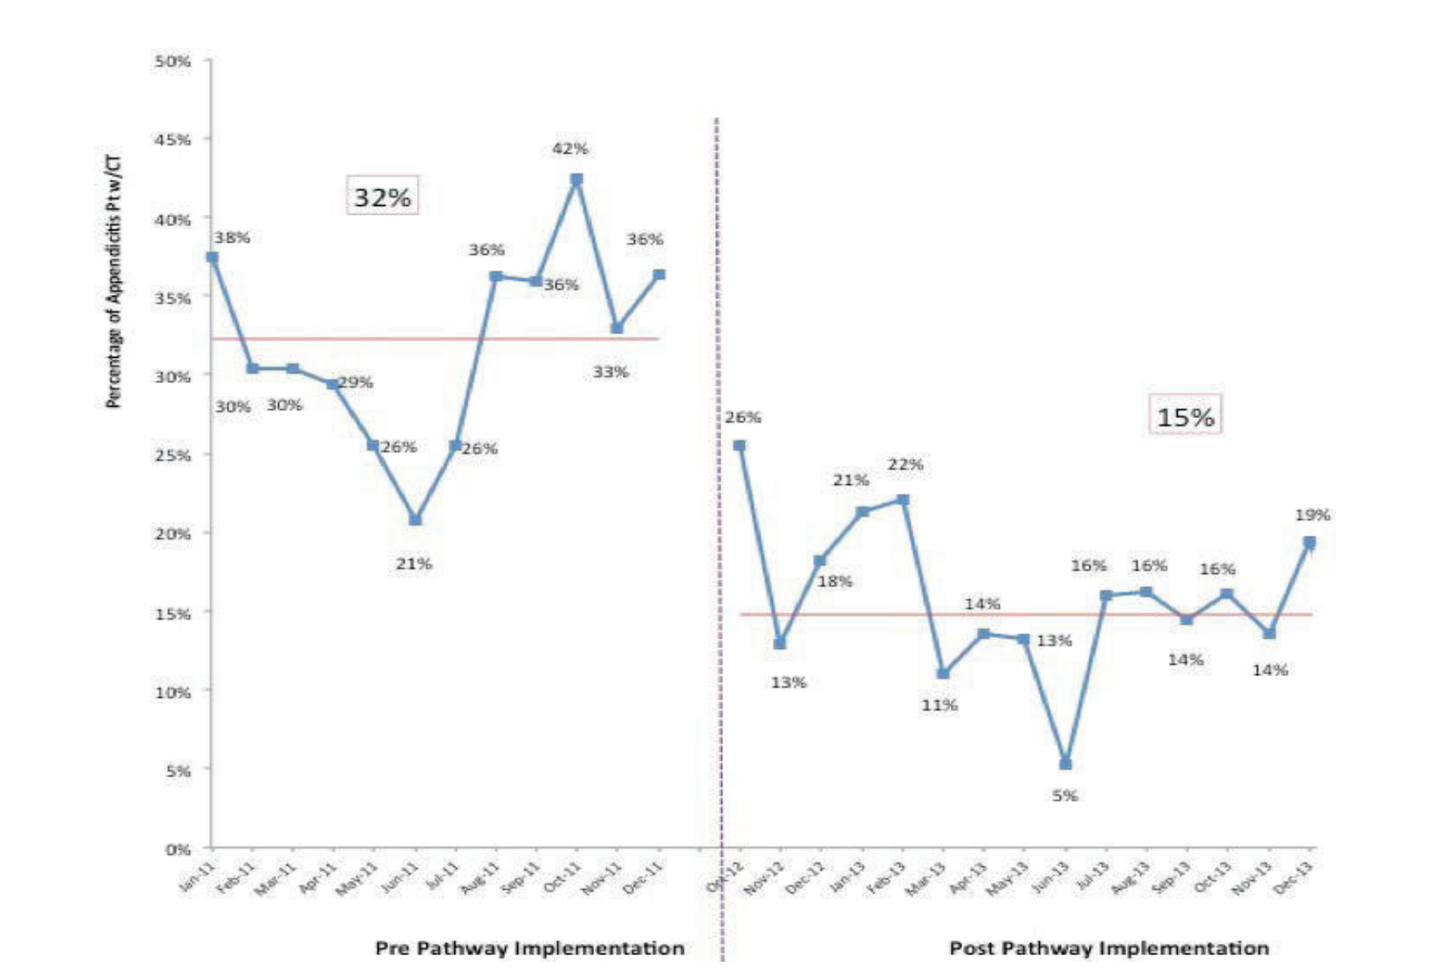

32%
15%
Percentage of Appendicitis Pt w/CT
Pre Pathway Implementation
Post Pathway Implementation

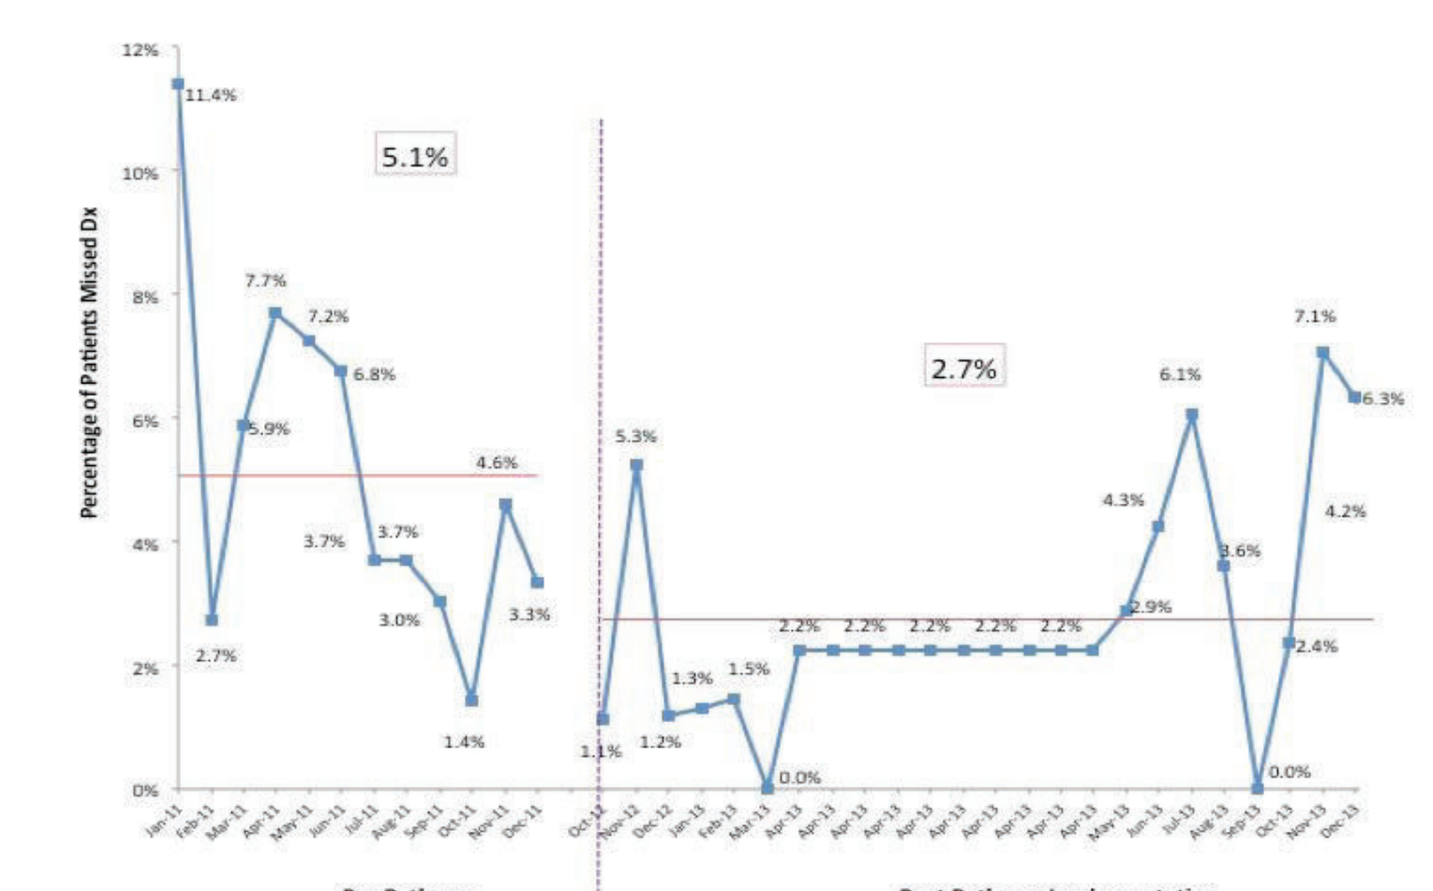

5.1%
2.7%
Percentage of Patients Missed Dx
Pre Pathway
Post Pathway Implementation

Figure 5.
Missed diagnosis rate of appendicitis difference between pre- and post-pathway implementation.

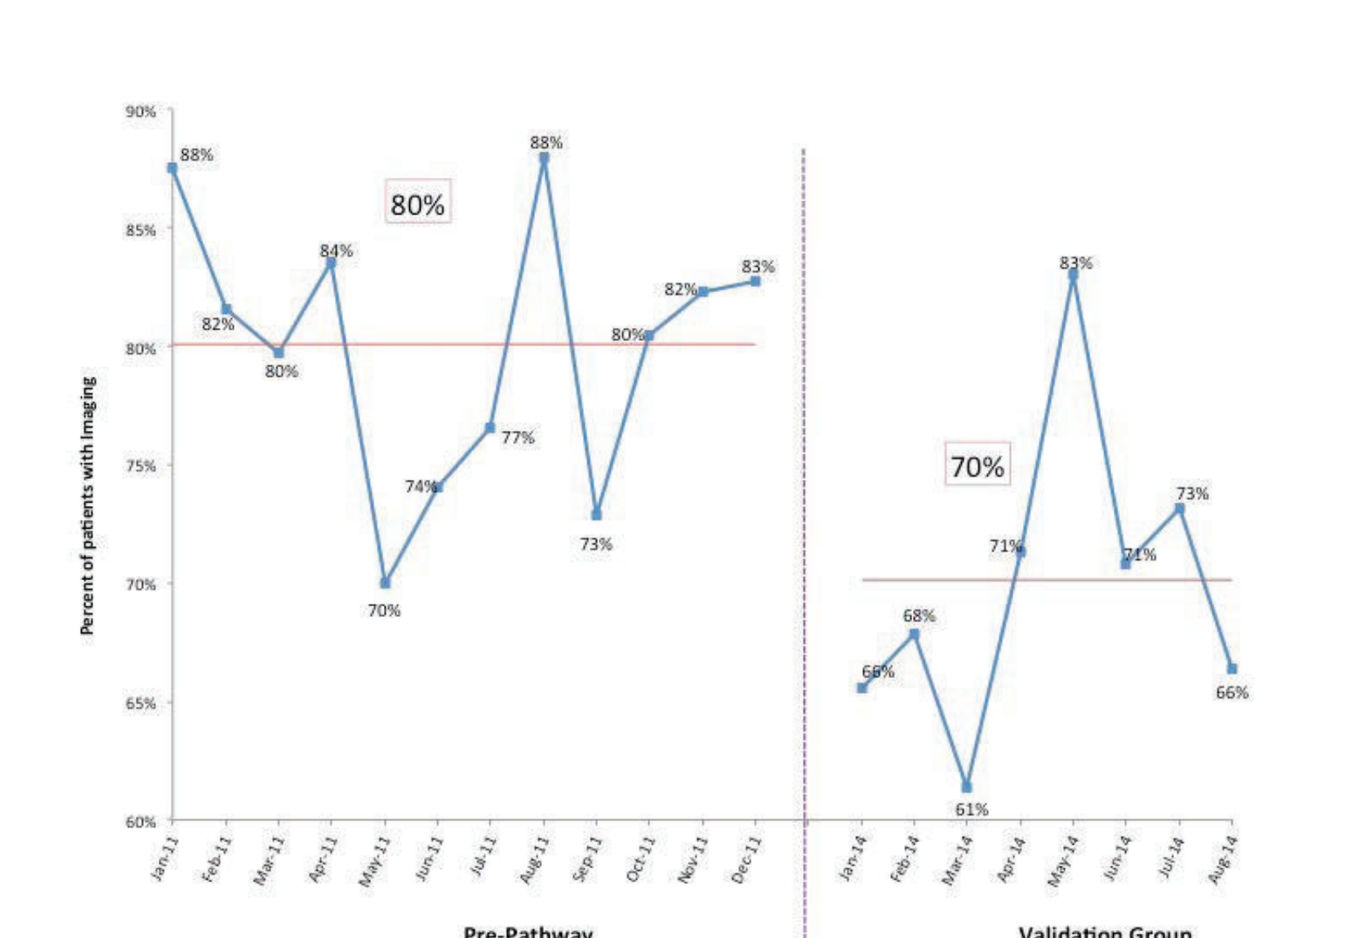

80%
70%
Percent of patients with imaging
Pre-Pathway
Validation Group

Figure 6..
Overall imaging utilization between pre-pathway implementation and validation groups.

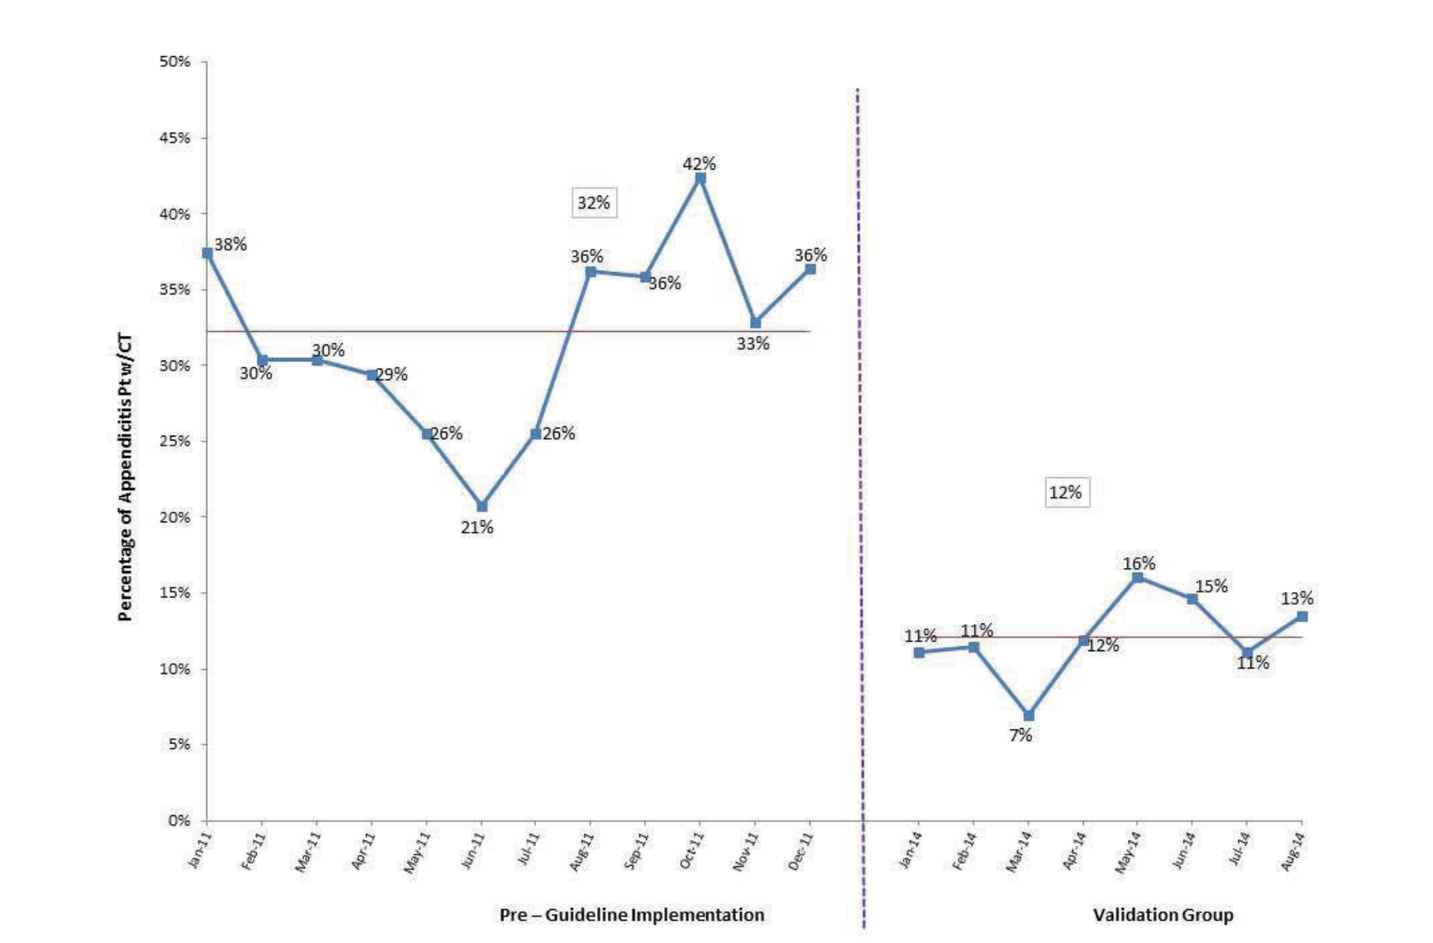

32%
12%
Percentage of Appendicitis Pt w/CT
Pre – Guideline Implementation
Validation Group

Figure 7.
CT utilization between pre-pathway implementation and validation groups.

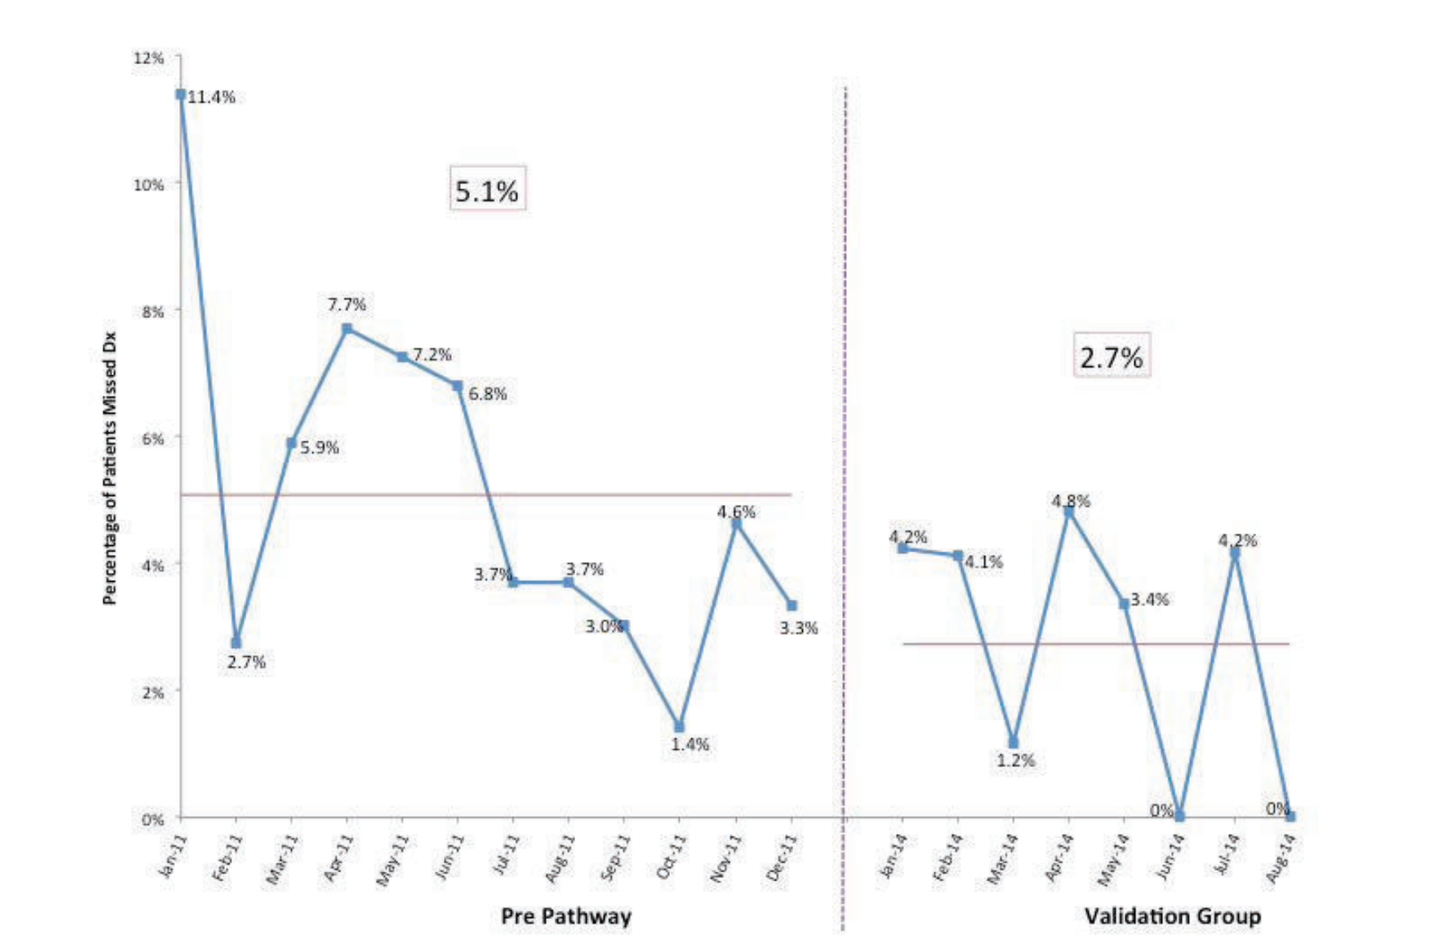

5.1%
2.7%
Percentage of Patients Missed Dx
Pre Pathway
Validation Group

Figure 8.
Missed diagnosis rate of appendicitis difference between pre-pathway implementation and validation groups.

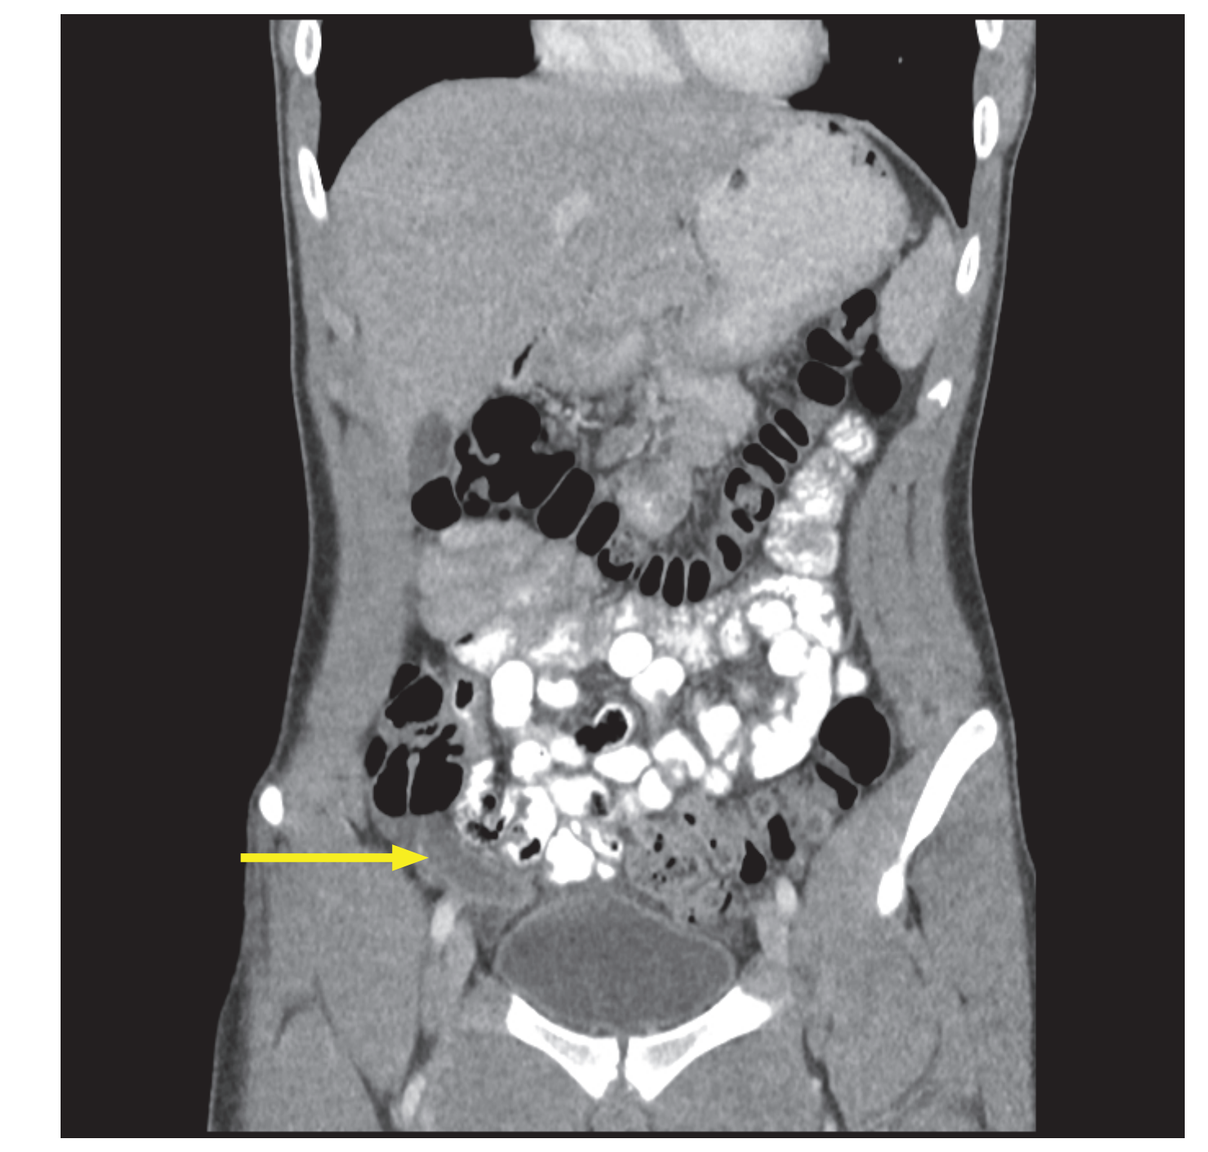

Figure 12.
Axial and coronal images demonstrating an enlarged appendix with surrounding inflammatory changes, consistent with diagnosis of appendicitis.

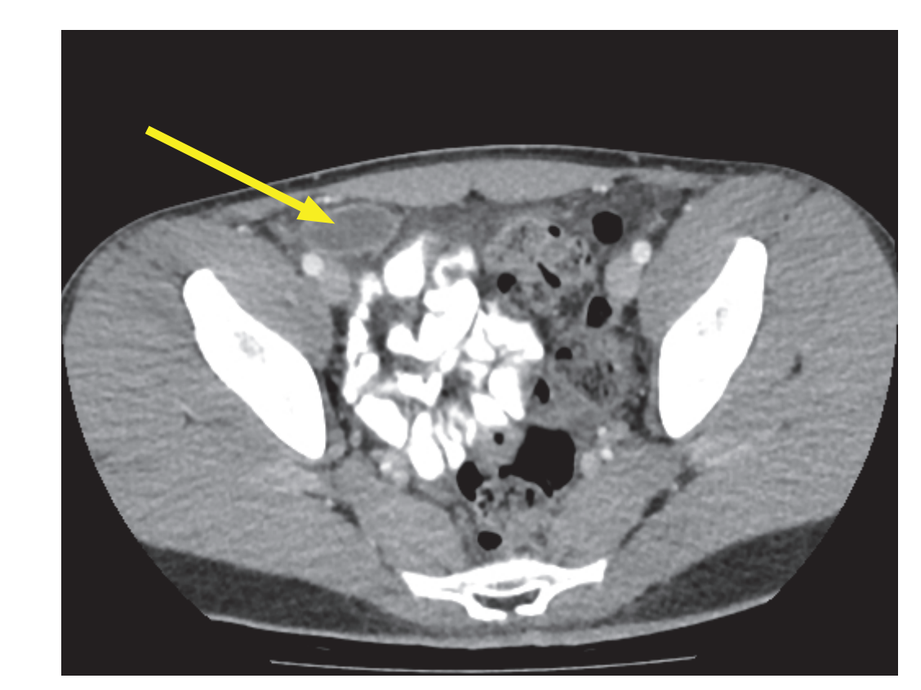

Table 1: Pre-pathway, Post-pathway and Validation Group Characteristics

	Pre Pathway	Percentage	Post Pathway	Percentage	Validation	Percentage	P value between Pre and Post Pathway	P value between Pre and Validation	P value between Post- Pathway and Validation
Number of patients	1079		1270		484				
Non perforated appendicitis	664	62%	763	60%	306	63%	0.47	0.54	0.25
Appendicitis with peritonitis	315	29%	366	29%	143	30%	0.85	0.9	0.77
Appendicitis with abscess formation	100	9%	141	11%	35	7%	0.15	0.21	0.06
CT or US Imaging	864	80%	902	71%	350	72%	0.001	0.001	0.64
US Imaging	683	63%	815	64%	315	65%	0.67	0.53	0.74
CT Imaging	348	32%	196	15%	69	14%	0.0001	0.0001	0.6
Missed Diagnosis	55	5%	39	3%	11	2%	0.02	0.01	0.42

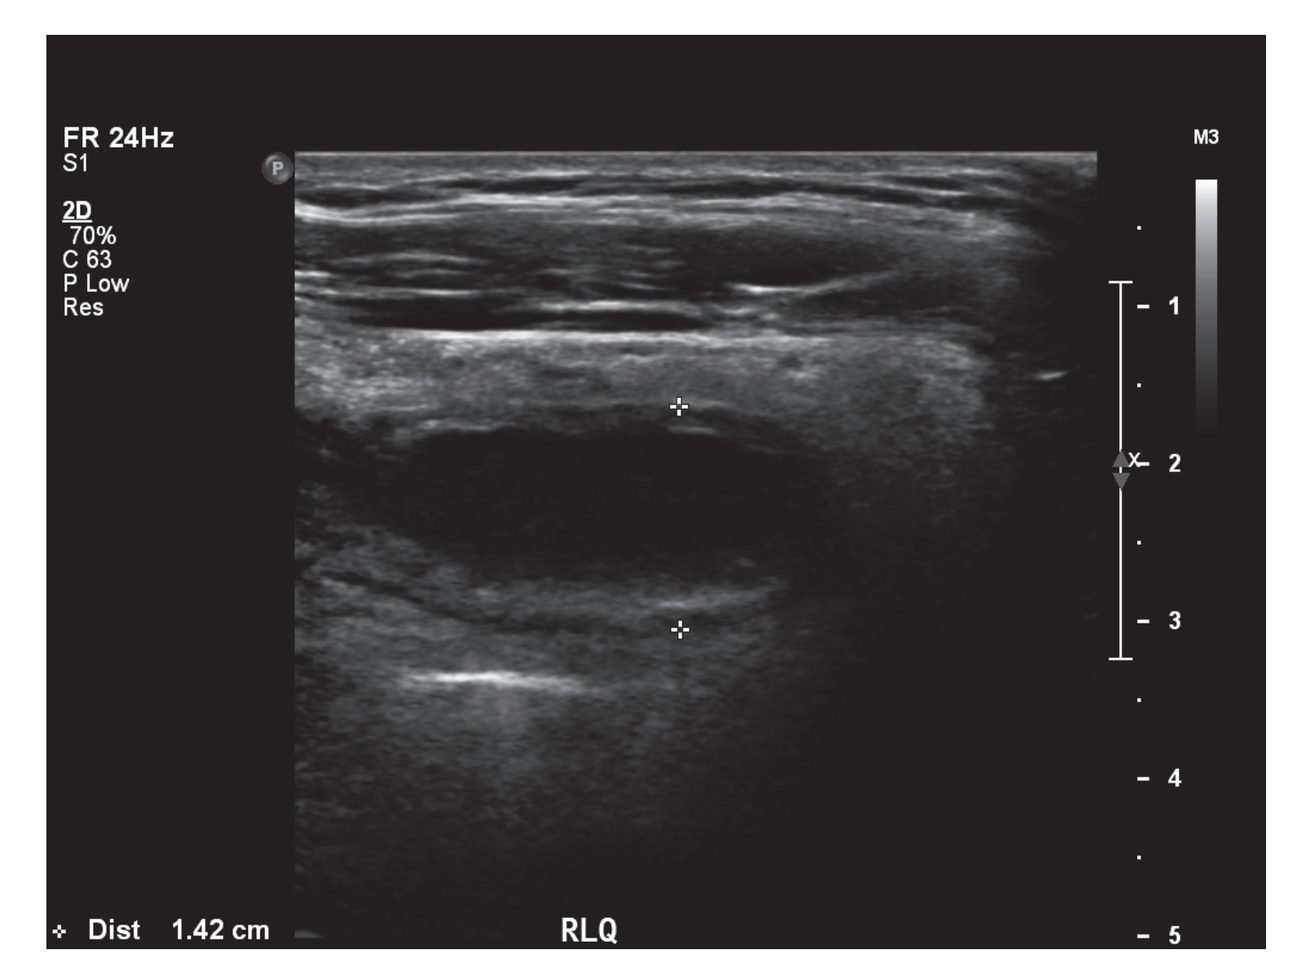

FR 24Hz
Dist 1.42 cm
RLQ

Figure 9.
Ultrasound imaging demonstrating dilated appendix, with surrounding echogenic fat, as seen in a patient with appendicitis.

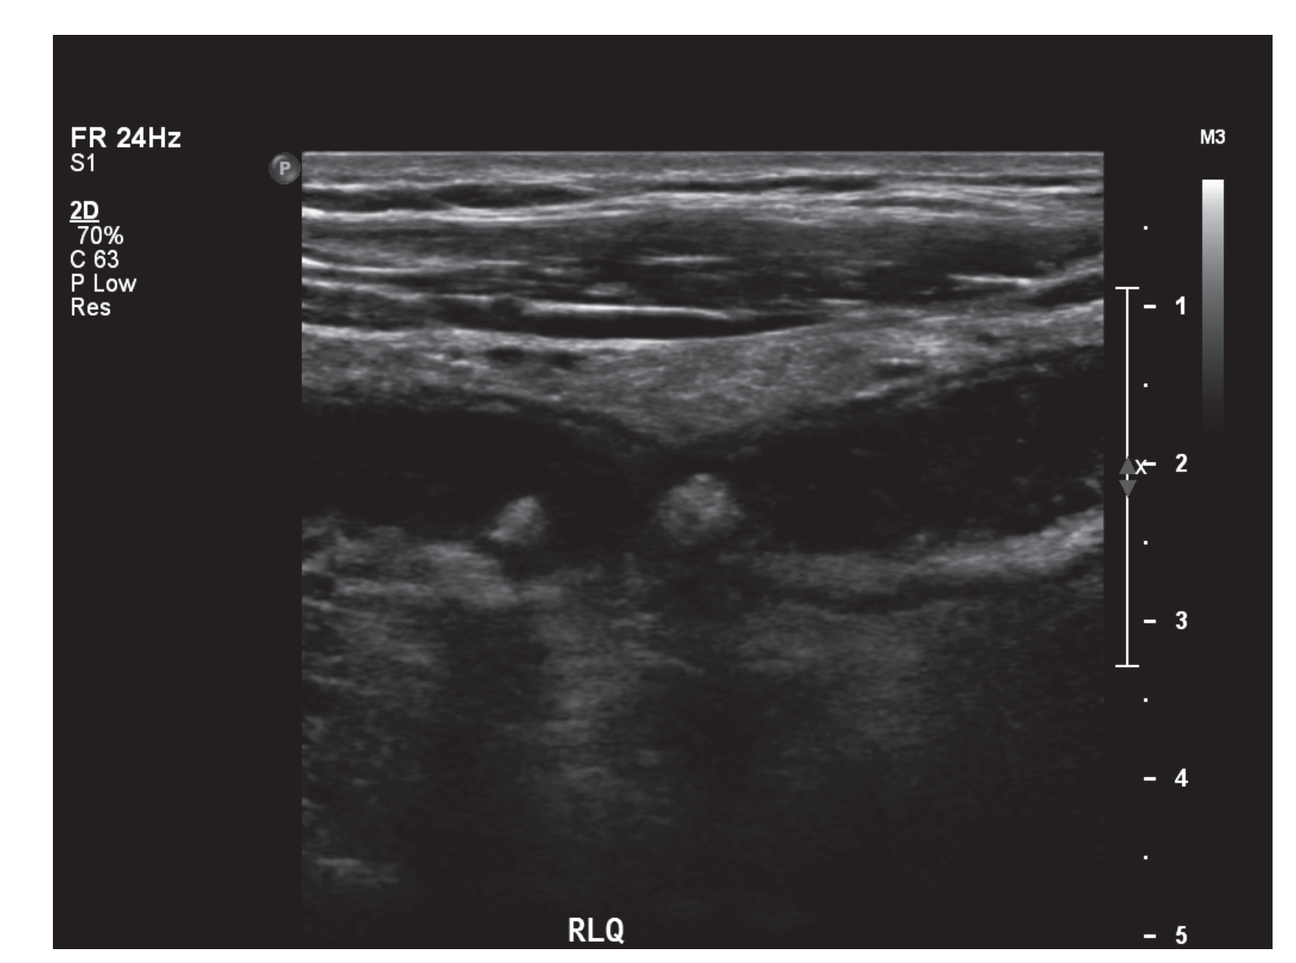

FR 24Hz
RLQ

Figure 10.
Ultrasound imaging demonstrating appendicolith within the appendix.

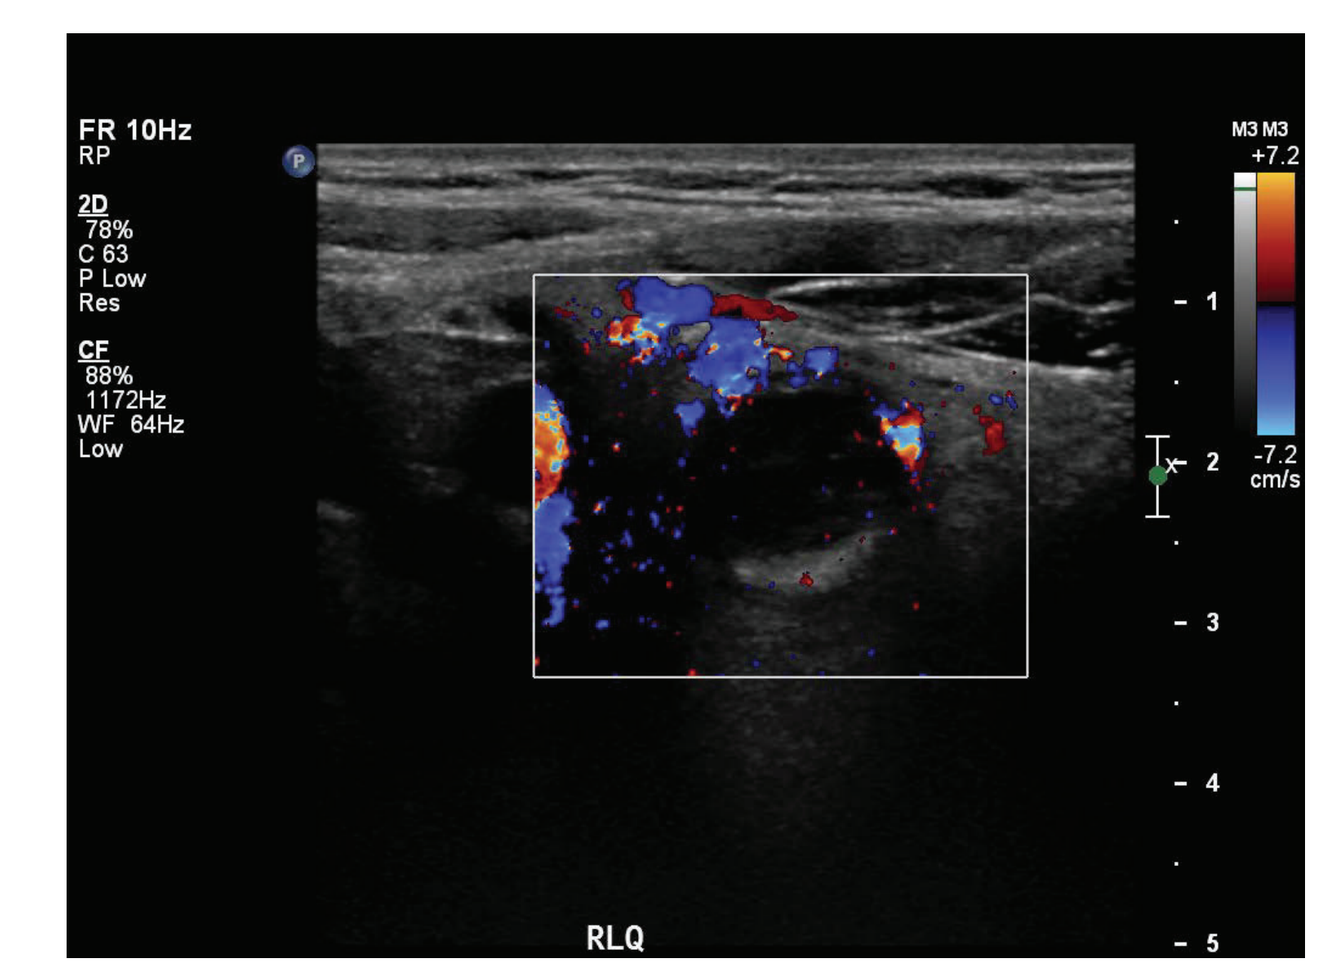

FR 10Hz
RLQ

Figure 11.
Hyperemia surrounding an enlarged appendix.

References

1. Goldman, R. D., et al. (2008). "Prospective Validation of the Pediatric Appendicitis Score." The Journal of pediatrics 153(2): 278-282.